FLUXION BIOSCIENCES

RESEARCH SPOTLIGHT

ELECTRICAL CHARACTERIZATION OF THE SHORT AND LONG GAMMA SUBUNIT SPLICE VARIANTS OF HUMAN GABA$_A$ RECEPTORS

Nathan Hunt[1], Ona McConnell[2], Eric Wang[2], Ali Yehia[1]

Abstract

Sleep aids, anti-anxiety medications, anesthetics, alcohol, and neurosteroids all target GABAA receptors (GABAARs), which regulate the bulk of rapid inhibition in the mammalian brain. GABAA receptors are pentamers of α, β and γ subunits, with the γ2 subunit having two splice variants (γ2S and γ2L). These two variants are distinguished by 8 amino acid differences when integrated into the pentameric receptors. It has been noted that they have assorted patterns of regional distribution during development and in the adult brain, with the γ2L dominating in later stages of life. Using a microfluidics-based automated patch clamp assay, we investigated the subtle ionic current differences between these γ2 splice variants under different conditions of agonist, allosteric modulation and block. Data will be presented from splice variants of human α1β2γ2, α2β2γ2 and α3β2γ2 GABA A receptors

Materials and Methods

Stable HEK-293 Cell lines expressing the different GABAA receptors were cultured at 37°C and 5% CO_2. Culture media composition is listed below.

Cells marked for experiments were kept in culture until they reached 65-75% confluence. Flasks were first washed with 10 ml of Ca2+ & Mg2+ PBS, followed by addition of Accutase®solution (Innovative Cell Technologies). After a brief incubation at 37°C for 3-5 minutes, the cell suspension was then spun down and re-suspended in 293 SFM II for at least 45 minutes (with gentle stirring).

Plates were water rinsed and filled with intracellular solutions and compound doses. DMSO was added to solutions not containing drugs to achieve a concentration of less than or equal to 0.2%. Right before the start of the experiment, cells were re-suspended in extracellular solution at a final concentration of 6 million/ml and placed in the plate cell inlet wells. Cells were continuously held at -60 mV. GABA was added for 3 seconds with a wash period of 20 seconds. All recordings were made with ensemble plates (20 cells) in the IonFlux system with whole cell patch clamp configuration.

Culture Media:

DMEM High Glucose w/pyruvate, 10% FBS, 1% Penicillin/ Streptomycin, 10 µg/ml Blasticidin, 130 µg/ml Hygromycin, 2 µg/ml Puromycin

Intracellular solution (mM):

KF 70, KCl 60, NaCl 15, HEPES 5, EGTA 5, MgATP 4, (pH 7.3)

Extracellular solution (mM):

NaCl 137, KCl 4, $MgCl_2$ 1, $CaCl_2$ 1.8, HEPES 10, Glucose 10 (pH 7.45)

GABA dose response (µM):

0.3, 1, 3, 10, 30, 100, 300

Diazepam dose response (µM):

0.01, 0.03 ,0.1 ,0.3 ,1 , 3

Rc-Ome dose response (µM):

0.1, 0.3, 1, 3, 10, 30

Picrotoxin dose response (µM):

0.3, 1, 3, 10, 30, 100

1.Fluxion Biosciences, Alameda, CA, USA

2.Dept. of Molecular Genetics & Microbiology, Center for Neurogenetics, University of Florida, Gainesville, FL, USA

IonFlux Mercury

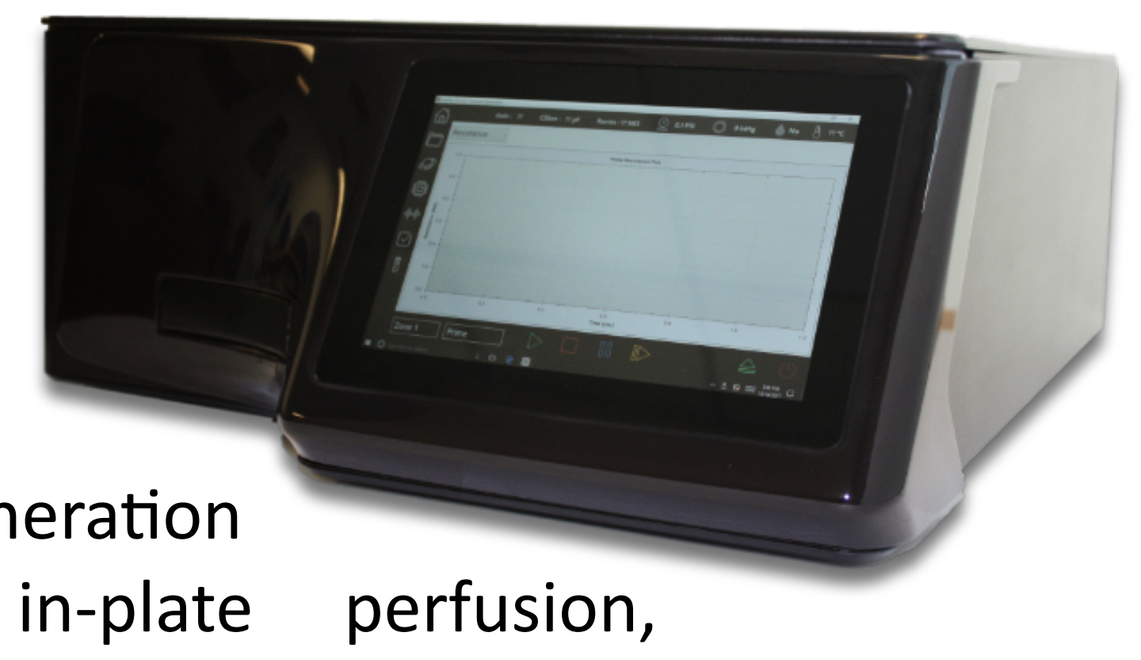

The IonFlux Mercury fully integrated automated patch clamp system features a next-generation pneumatically driven in-plate perfusion, providing precise fluid control and continuous flow.

IonFlux plates, based on standard configurations of 96 or 384 wells can be preloaded with cells, compounds, and reagents using conventional liquid handlers or pipettors. Each pattern in an IonFlux plate has 8 compound wells, 2 trap recordings filled with intracellular fluid, 1 inlet for cells and extracellular solution, and 1 outlet for waste (below). Cells are patched in whole cell configuration. Plates come in two configurations: 20-cell ensemble and single cell. Both plate formats provide high quality recordings, with giga-seals recorded in single-cell plates. Currents displayed here were recorded in ensemble format.

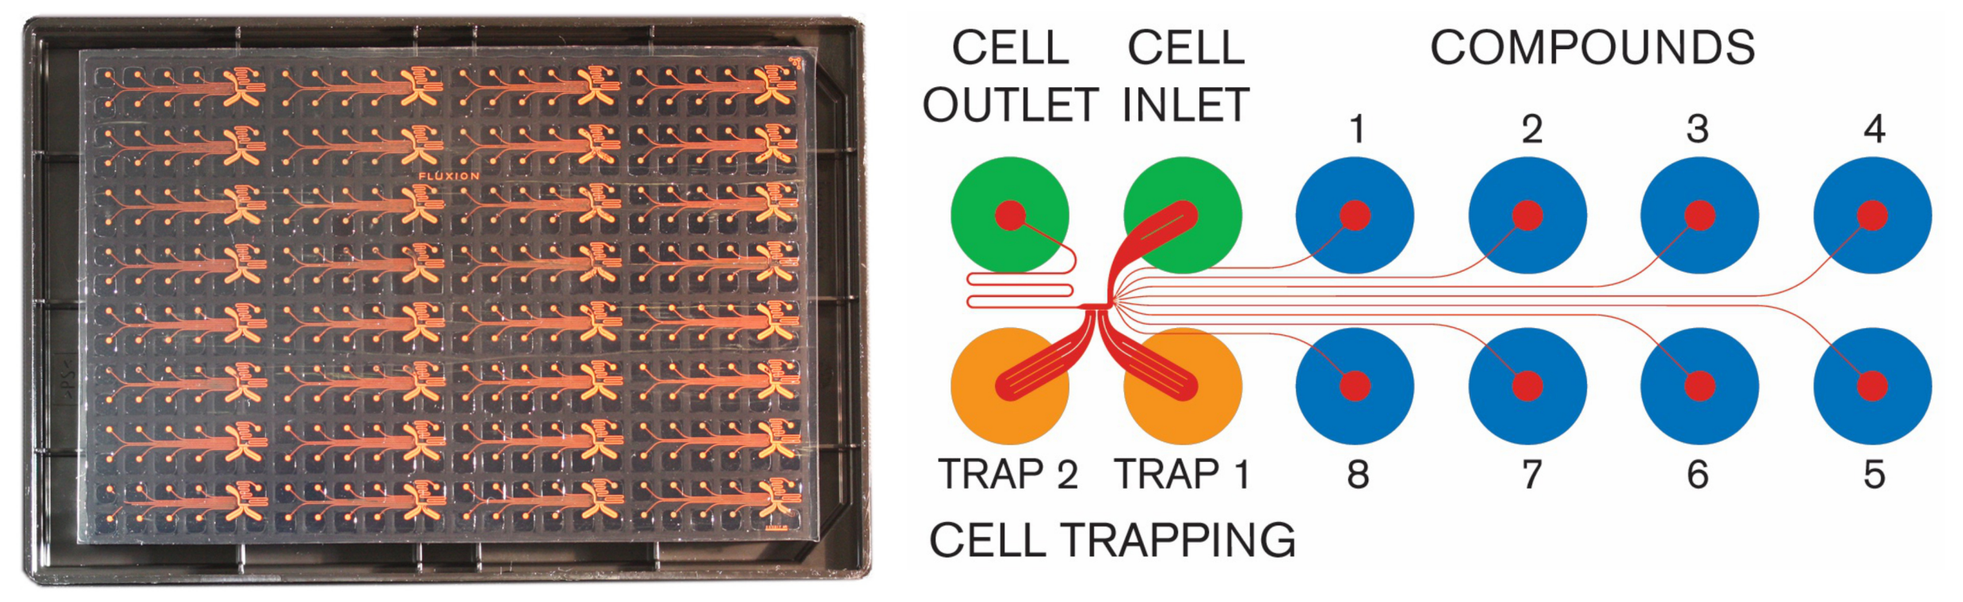

CELL OUTLET
CELL INLET
COMPOUNDS
1 2 3 4
TRAP 2 TRAP 1
8 7 6 5
CELL TRAPPING

GABA Response

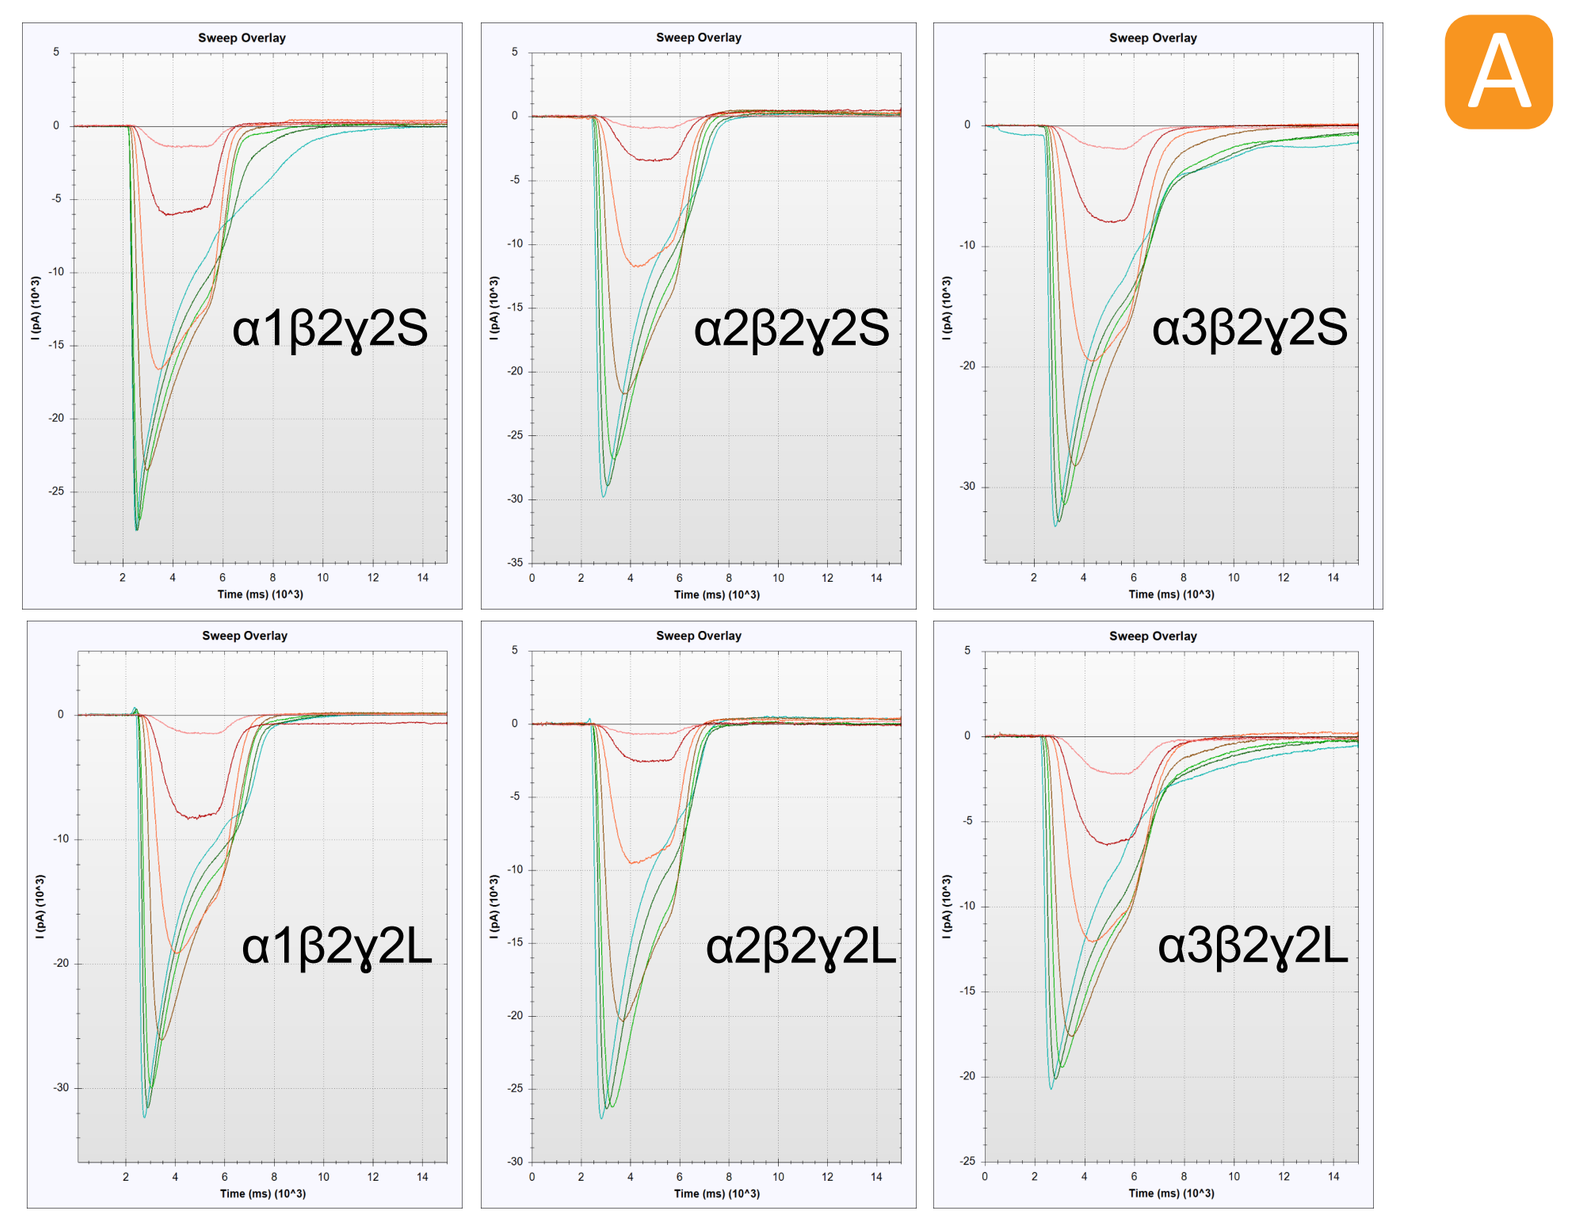

A
α1β2γ2S
α2β2γ2S
α3β2γ2S
α1β2γ2L
α2β2γ2L
α3β2γ2L

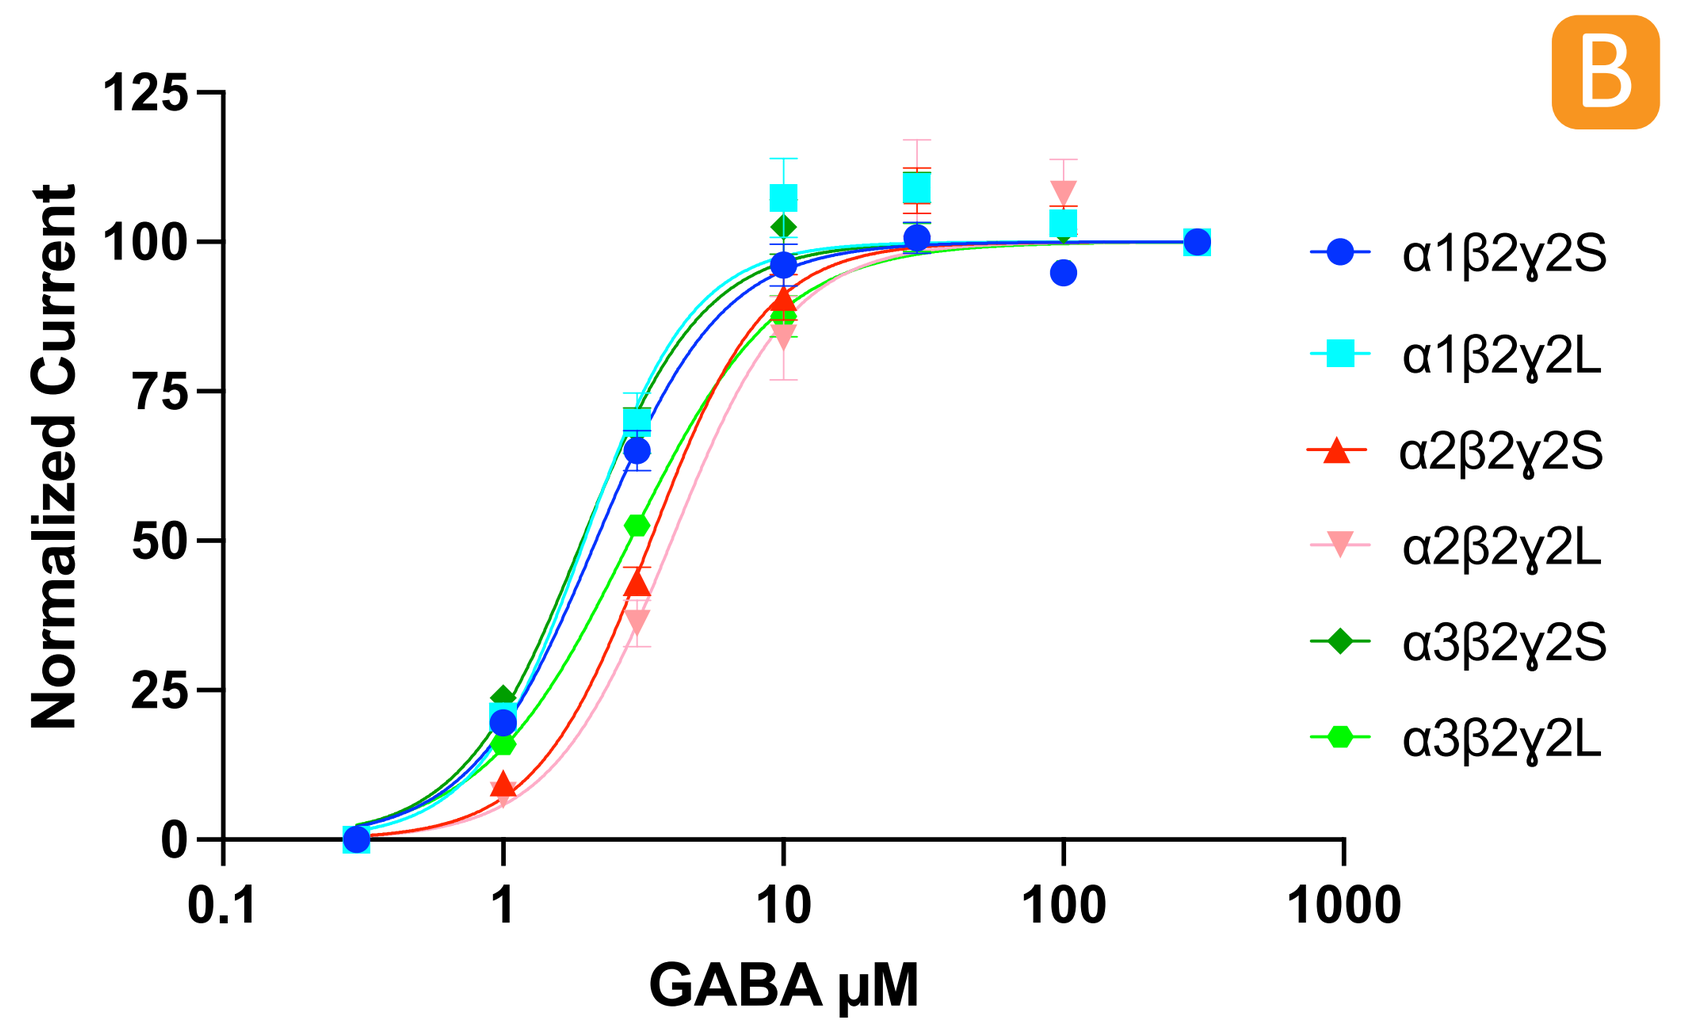

B
Normalized Current
GABA µM
α1β2γ2S
α1β2γ2L
α2β2γ2S
α2β2γ2L
α3β2γ2S
α3β2γ2L

GABA dose responses were created by sequentially adding an increasing GABA concentration. **A:** Representative superimposed sweeps from every cell line showing the increasing GABA-induced current. **B:** Normalized dose response curve for every cell line (n=8). Error bars represent SEM. EC_{50} values are displayed in the table.

Effect of Diazepam & Rc-Ome

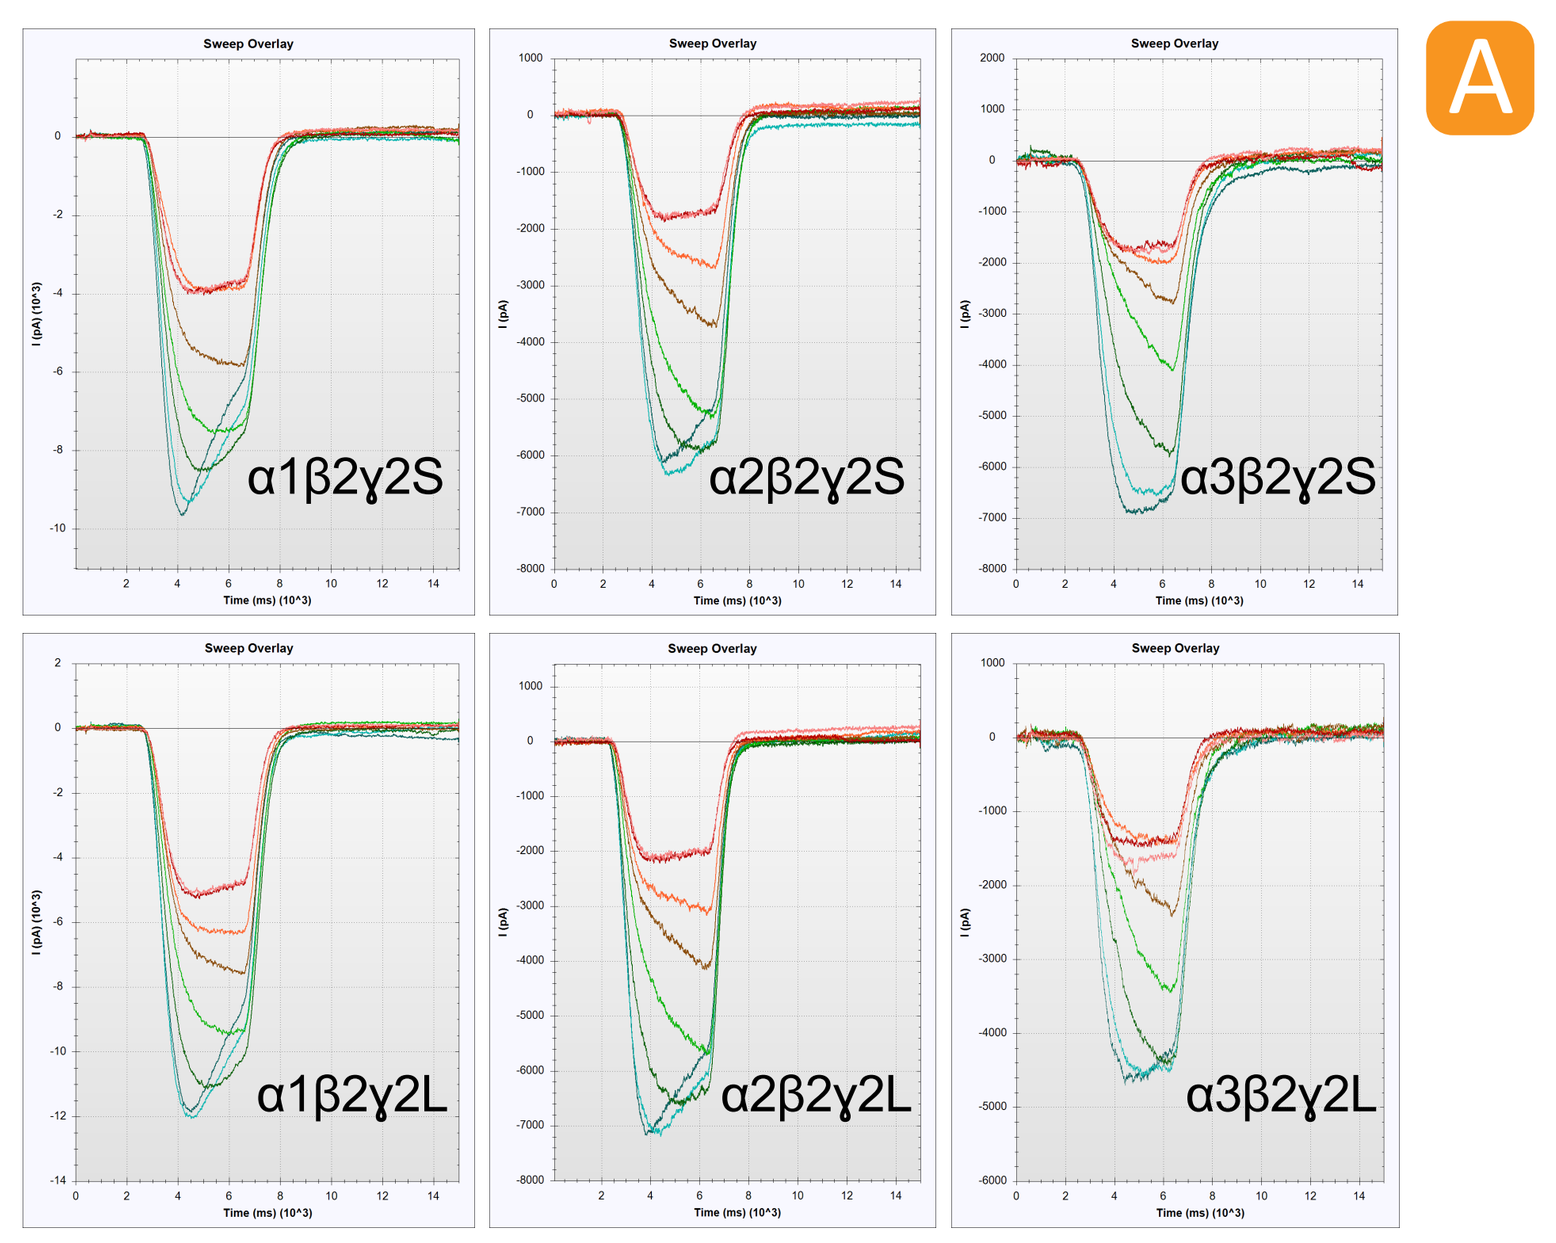

A
α1β2γ2S
α2β2γ2S
α3β2γ2S
α1β2γ2L
α2β2γ2L
α3β2γ2L

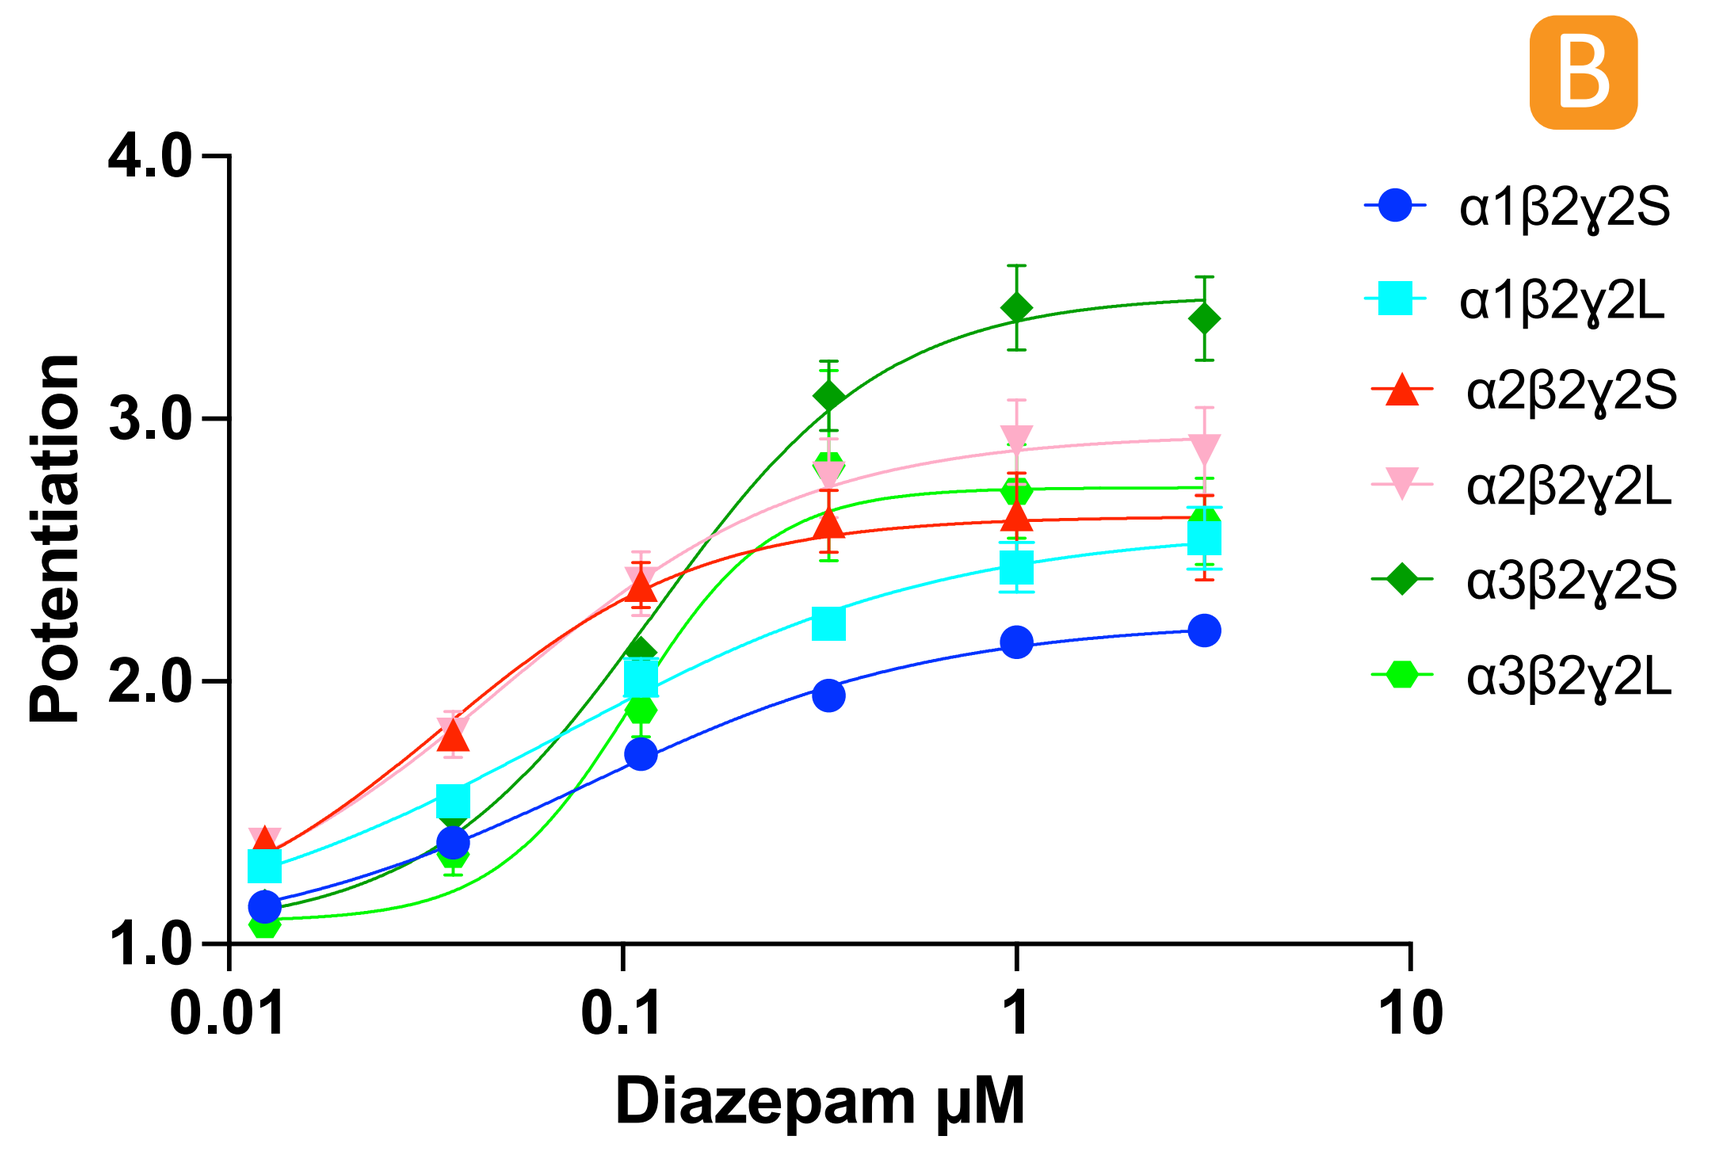

B
Potentiation
Diazepam µM
α1β2γ2S
α1β2γ2L
α2β2γ2S
α2β2γ2L
α3β2γ2S
α3β2γ2L

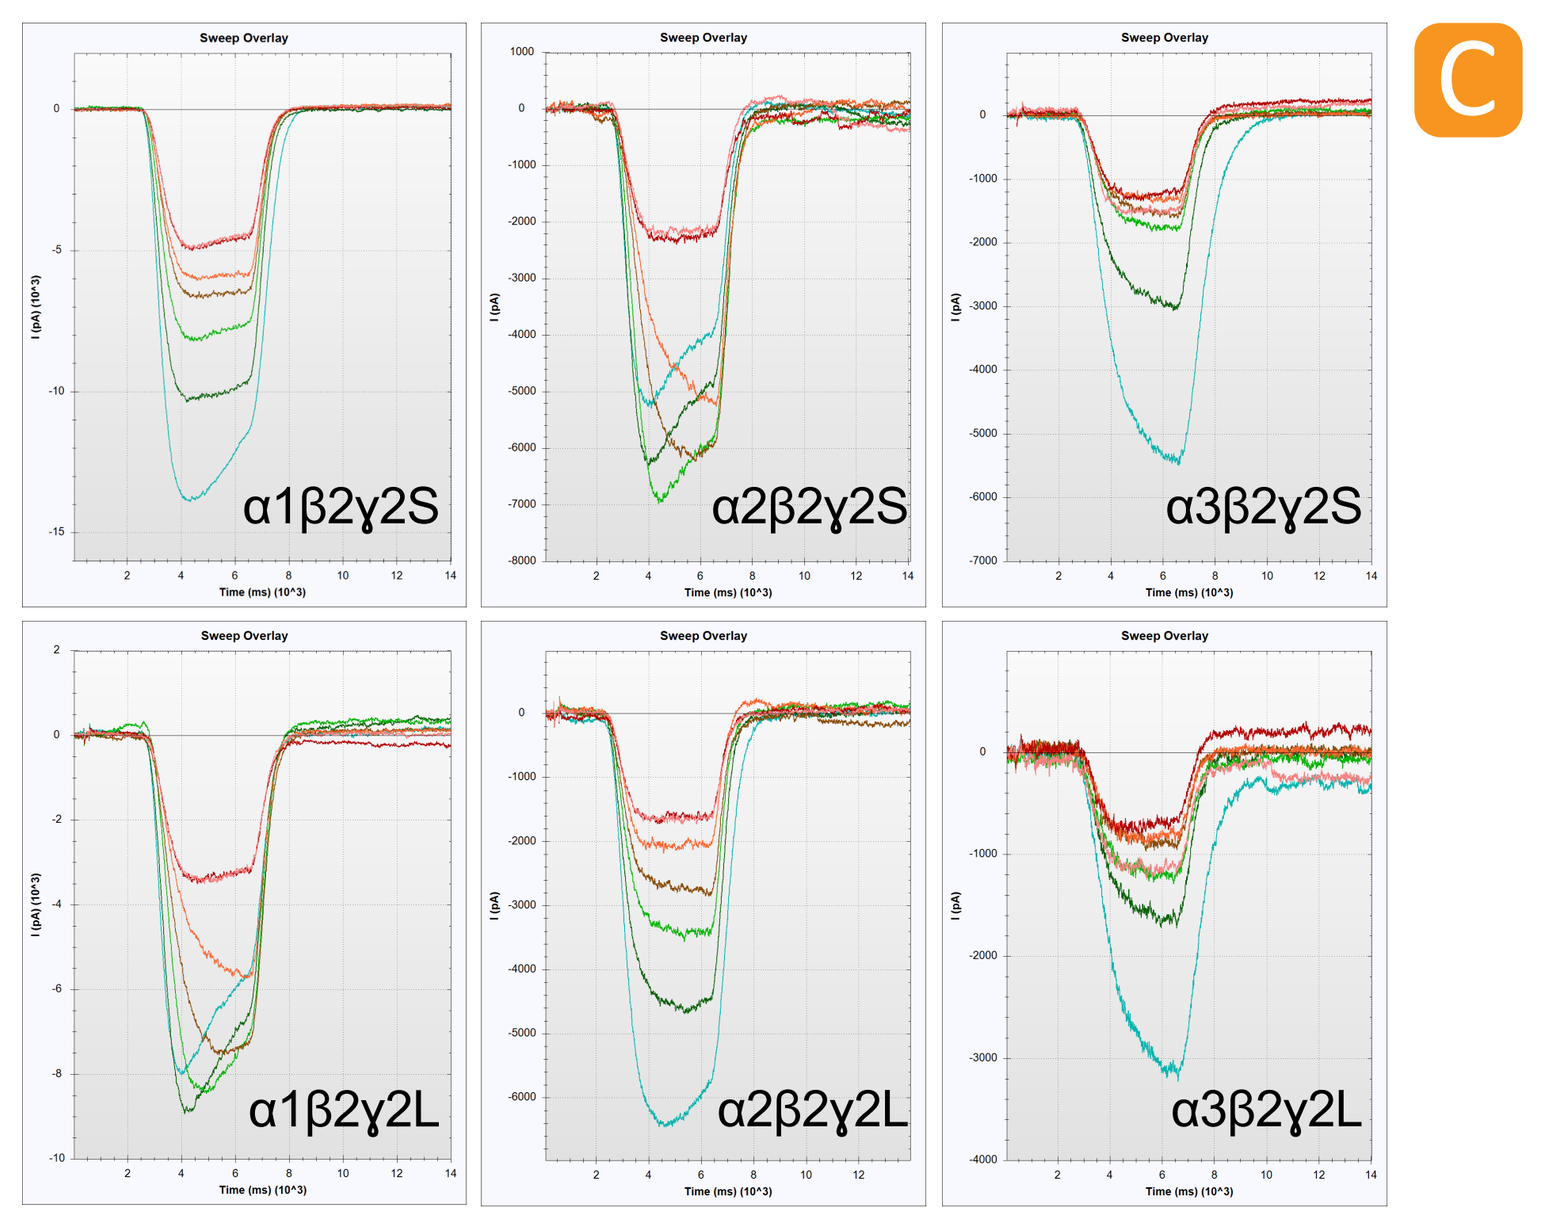

C
α1β2γ2S
α2β2γ2S
α3β2γ2S
α1β2γ2L
α2β2γ2L
α3β2γ2L

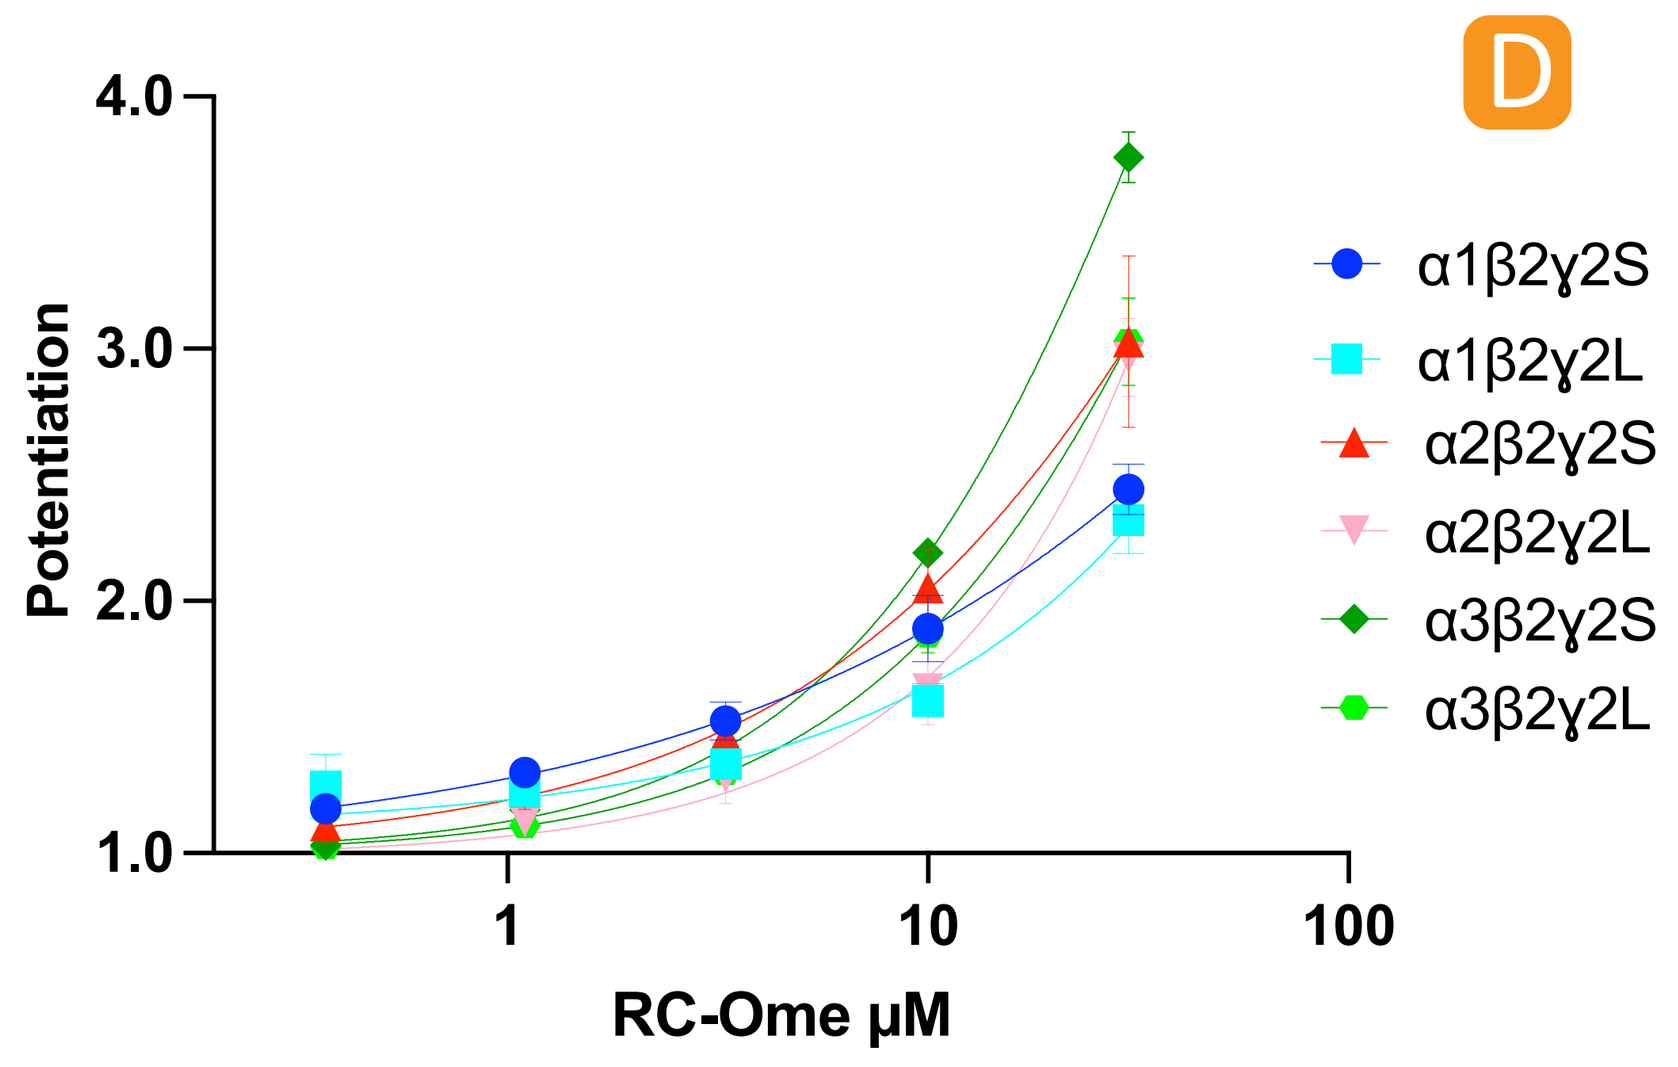

D
Potentiation
RC-Ome µM
α1β2γ2S
α1β2γ2L
α2β2γ2S
α2β2γ2L
α3β2γ2S
α3β2γ2L

Diazepam and Rc-Ome dose responses were created by sequentially adding an increasing PAM concentration with 1 µM GABA. **A:** Representative superimposed sweeps from every cell line showing diazepam potentiation. **B:** Potentiation dose response curve for every cell line (n=6). **C:** Representative superimposed sweeps showing Rc-Ome potentiation. **D:** Potentiation dose response curve for every cell line (n=6). Error bars represent SEM. EC_{50} values for diazepam are displayed in the table. Values for Rc-Ome could not be calculated.

Effect of Picrotoxin

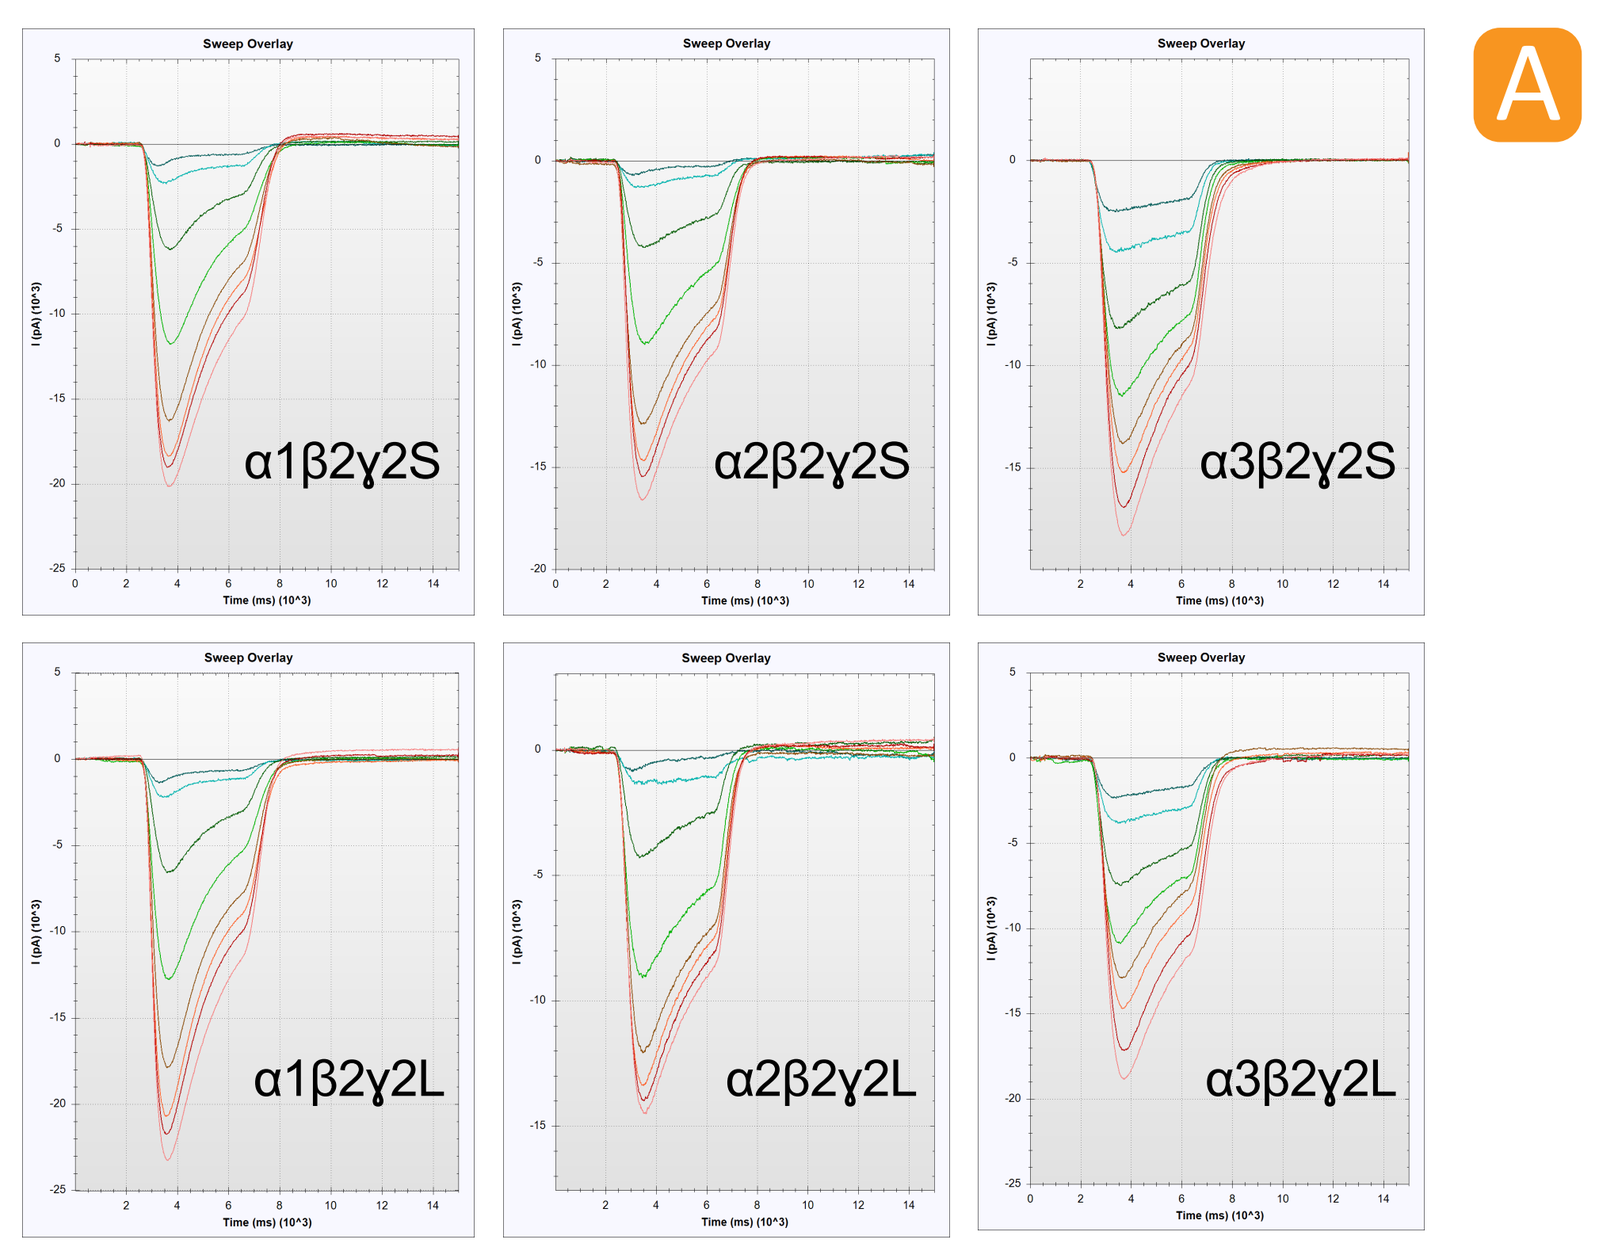

A
α1β2γ2S
α2β2γ2S
α3β2γ2S
α1β2γ2L
α2β2γ2L
α3β2γ2L

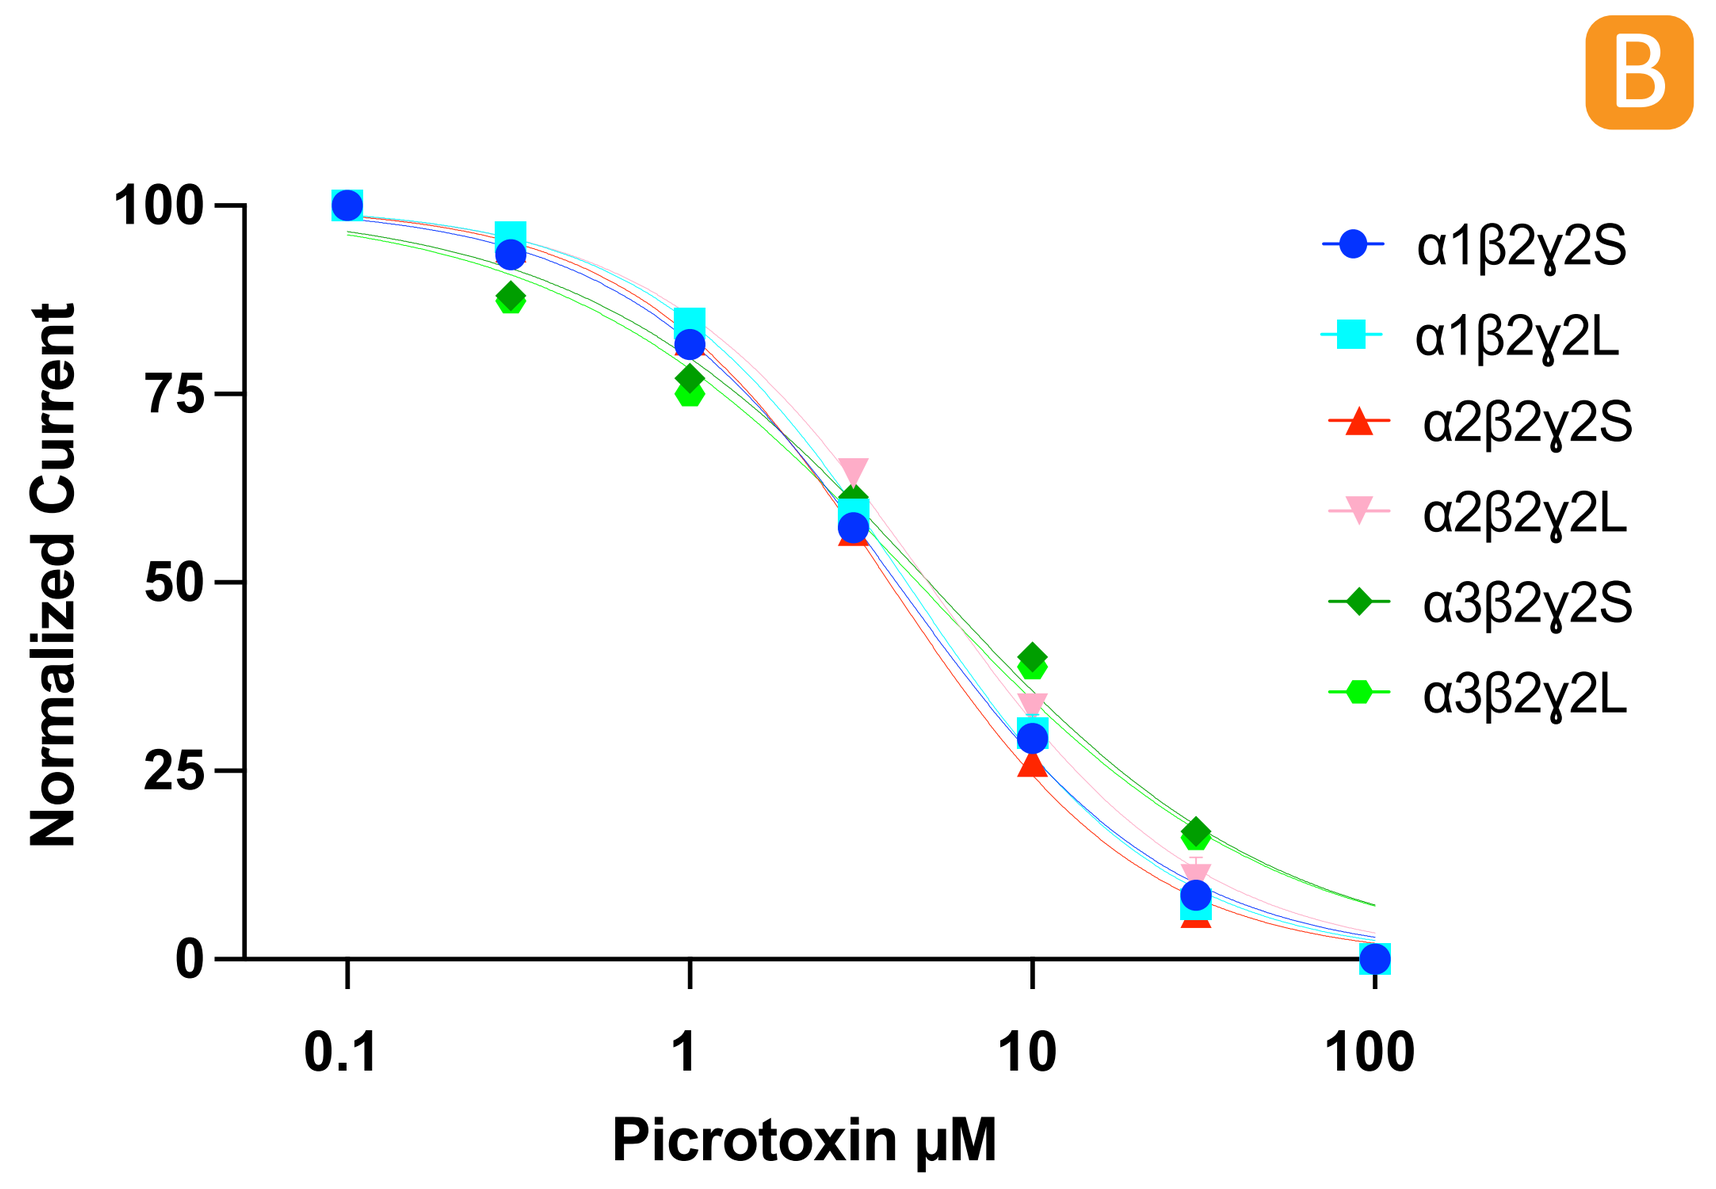

B
Normalized Current
Picrotoxin µM
α1β2γ2S
α1β2γ2L
α2β2γ2S
α2β2γ2L
α3β2γ2S
α3β2γ2L

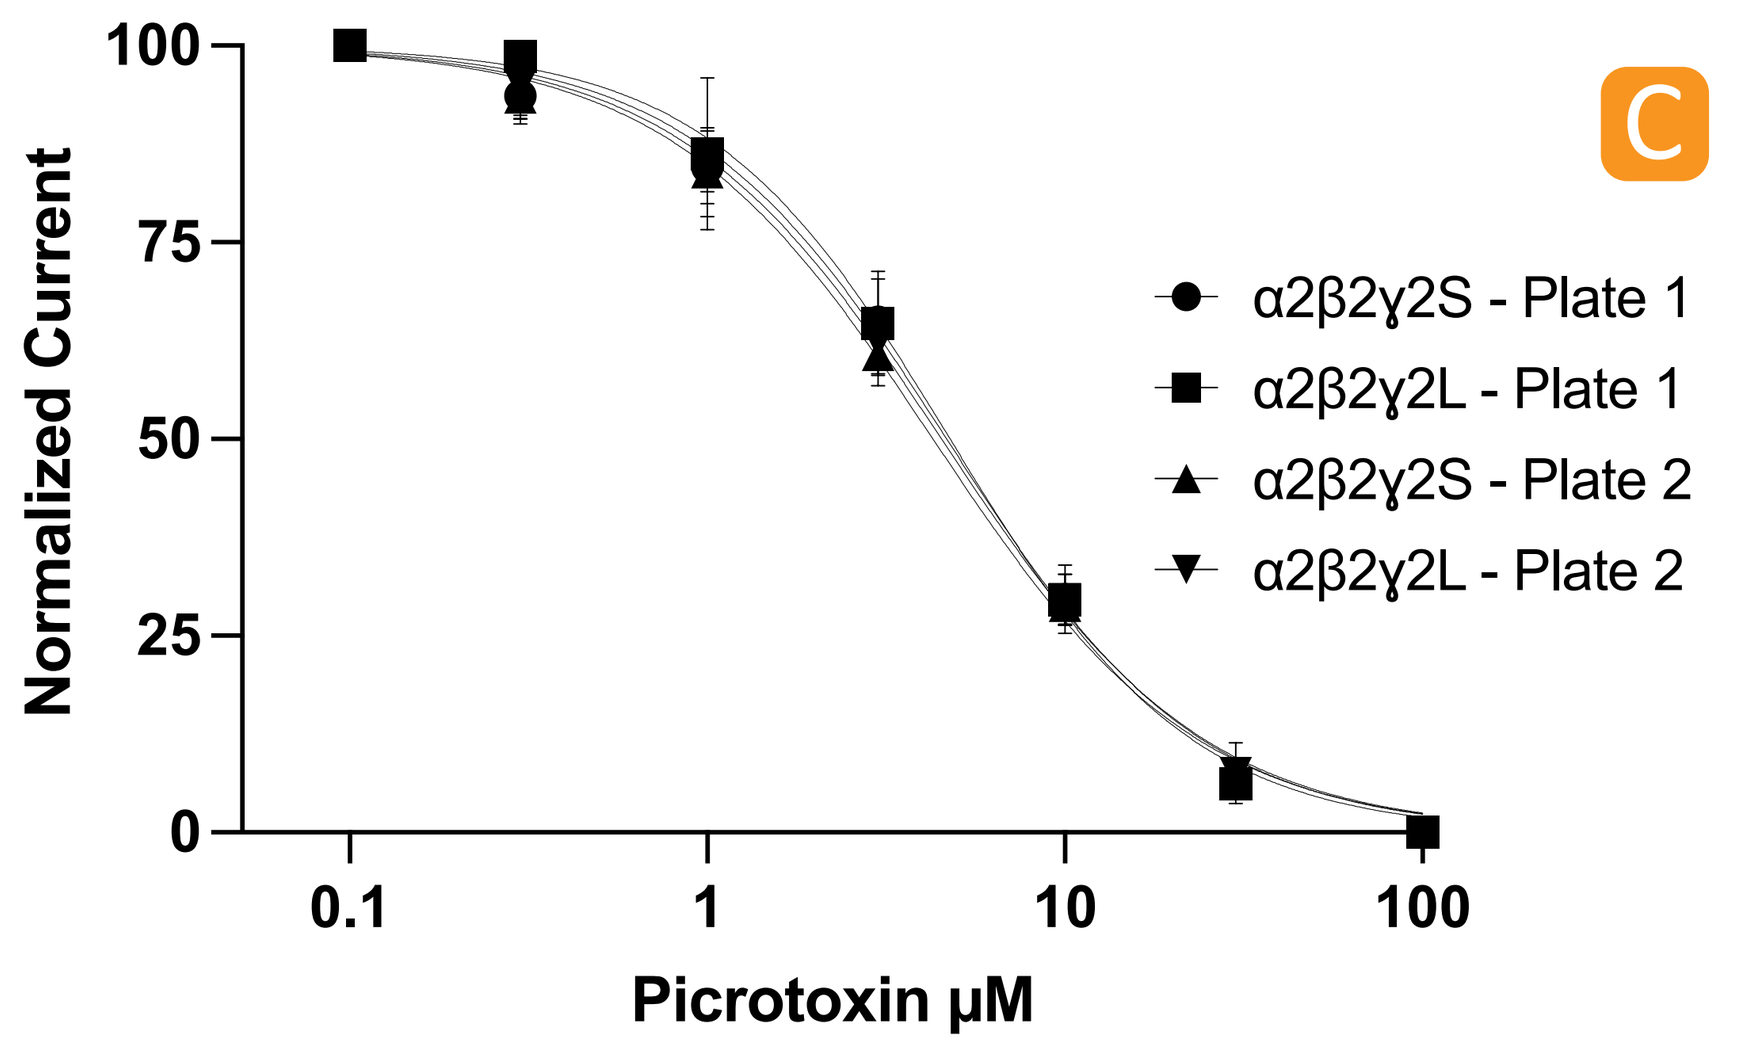

C
Normalized Current
Picrotoxin µM
α2β2γ2S - Plate 1
α2β2γ2L - Plate 1
α2β2γ2S - Plate 2
α2β2γ2L - Plate 2

Picrotoxin dose responses were created by sequentially adding an increasing antagonist concentration with 10 µM GABA. **A:** Representative superimposed sweeps from every cell line showing picrotoxin inhibition. **B:** Normalized inhibition dose response curve for every cell line (n=8). **C:** Superimposed picrotoxin inhibition dose response curves from two different plates showing the reproducibility of the results. Error bars represent SEM. IC_{50} in µM are displayed in the table.

µM	α1β2γ2S	α1β2γ2L	α2β2γ2S	α2β2γ2L	α3β2γ2S	α3β2γ2L
GABA	2.1	1.9	3.4	3.9	1.9	2.8
Diazepam	0.082	0.070	0.038	0.050	0.12	0.11
Picrotoxin	4.0	4.3	3.8	4.9	4.9	4.8

Reference

1. Jiang, R. et al. Retrochalcone derivatives are positive allosteric modulators at synaptic and extrasynaptic GABAA receptors in vitro. Brit J Pharmacol 162, 1326–1339 (2011).
2. Baburin, I. et al. Estimating the efficiency of benzodiazepines on GABAA receptors comprising γ1 or γ2 subunits. Brit J Pharmacol 155, 424–433 (2008).
3. Benkwitz, C., Banks, M. I. & Pearce, R. A. Influence of GABAA Receptor γ2 Splice Variants on Receptor Kinetics and Isoflurane Modulation. Anesthesiology.
4. Hosie, A. M., Dunne, E. L., Harvey, R. J. & Smart, T. G. Zinc-mediated inhibition of GABAA receptors: discrete binding sites underlie subtype specificity. Nat Neurosci 6, 362–369 (2003).
5. Boileau, A. J., Pearce, R. A. & Czajkowski, C. The Short Splice Variant of the 2 Subunit Acts as an External Modulator of GABAA Receptor Function. J Neurosci 30, 4895–4903 (2010).

Effect of $ZnCl_2$

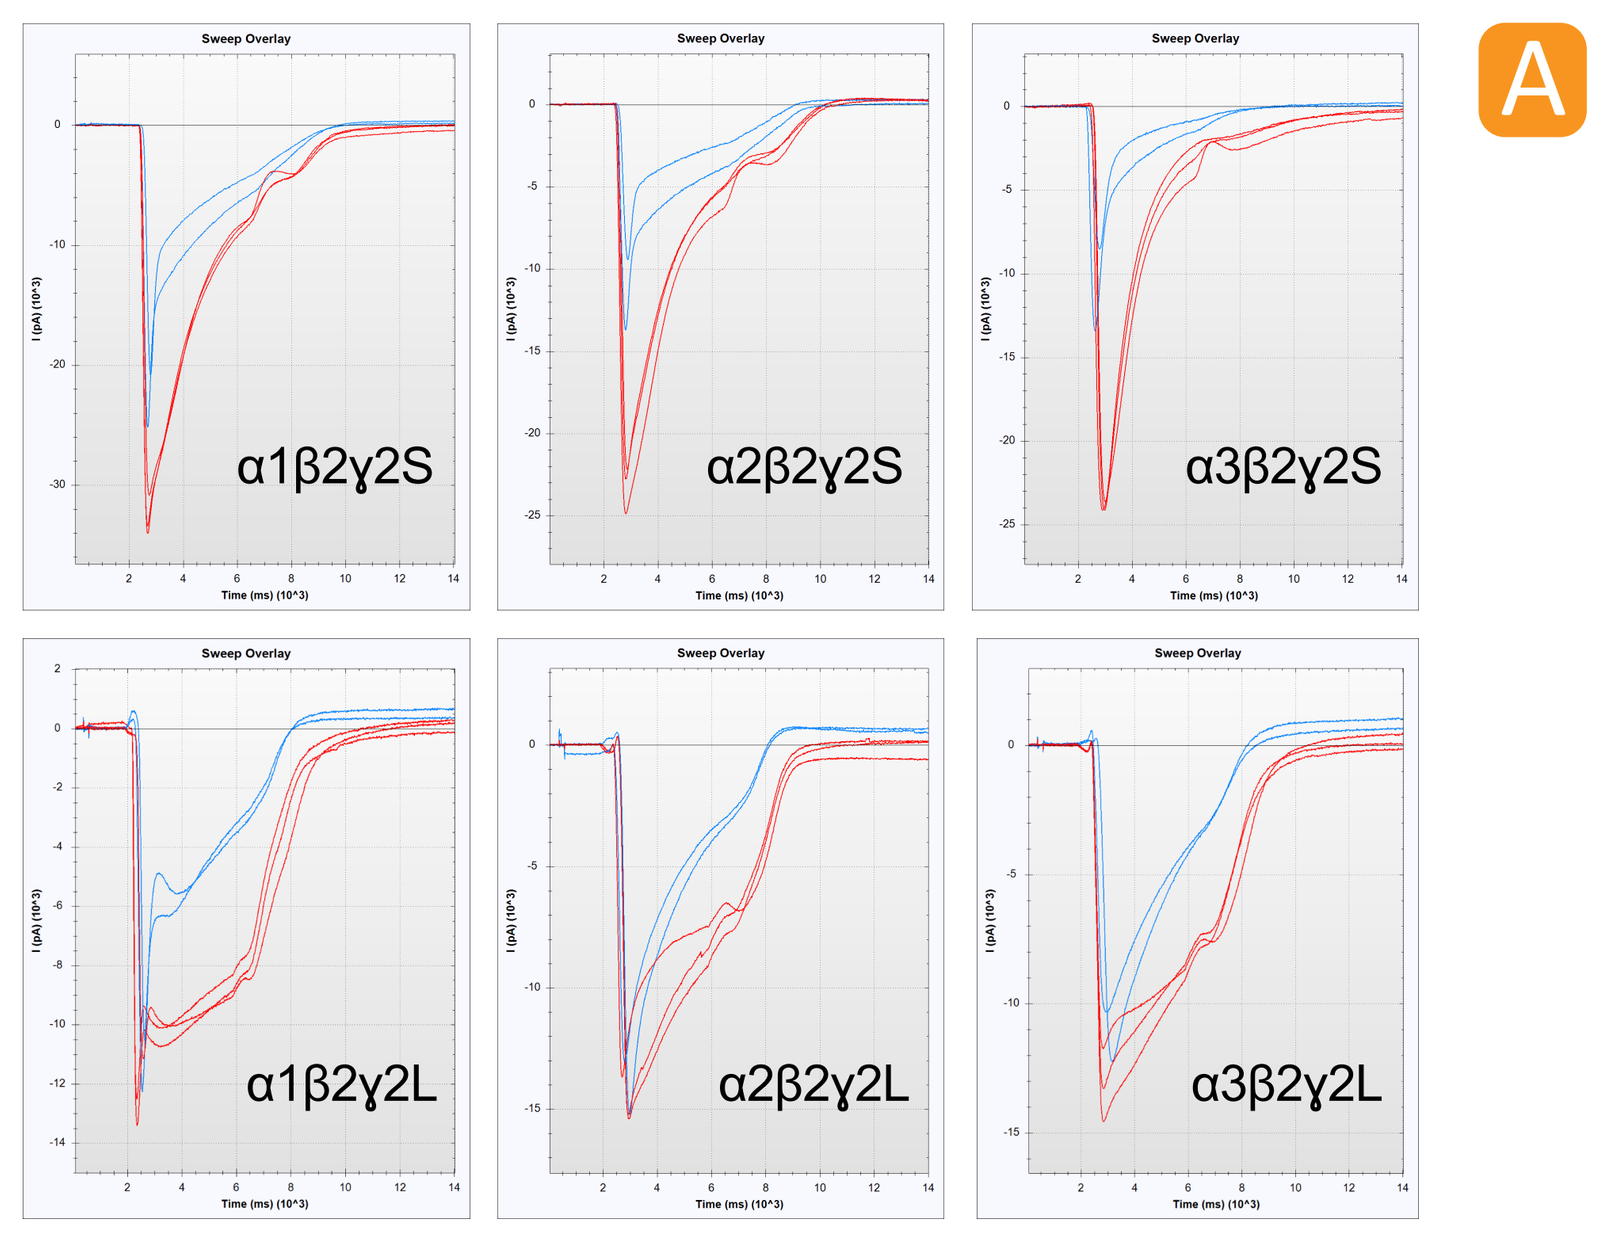

A
α1β2γ2S
α2β2γ2S
α3β2γ2S
α1β2γ2L
α2β2γ2L
α3β2γ2L

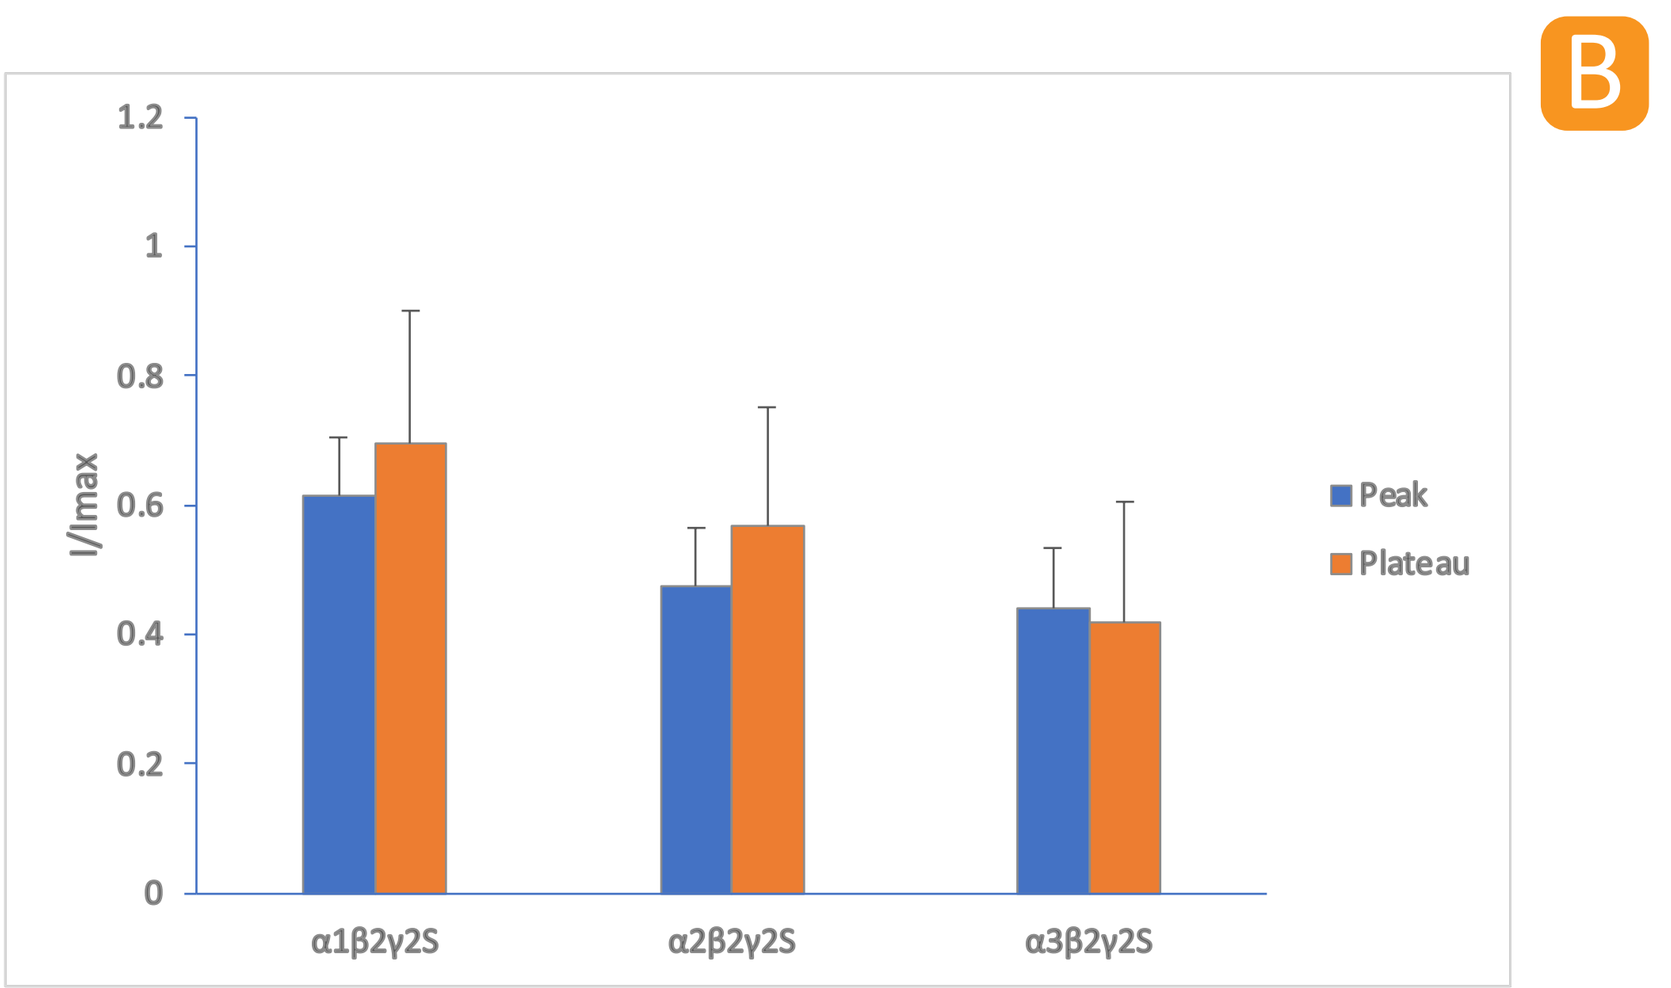

B
I/Imax
Peak
Plateau
α1β2γ2S
α2β2γ2S
α3β2γ2S

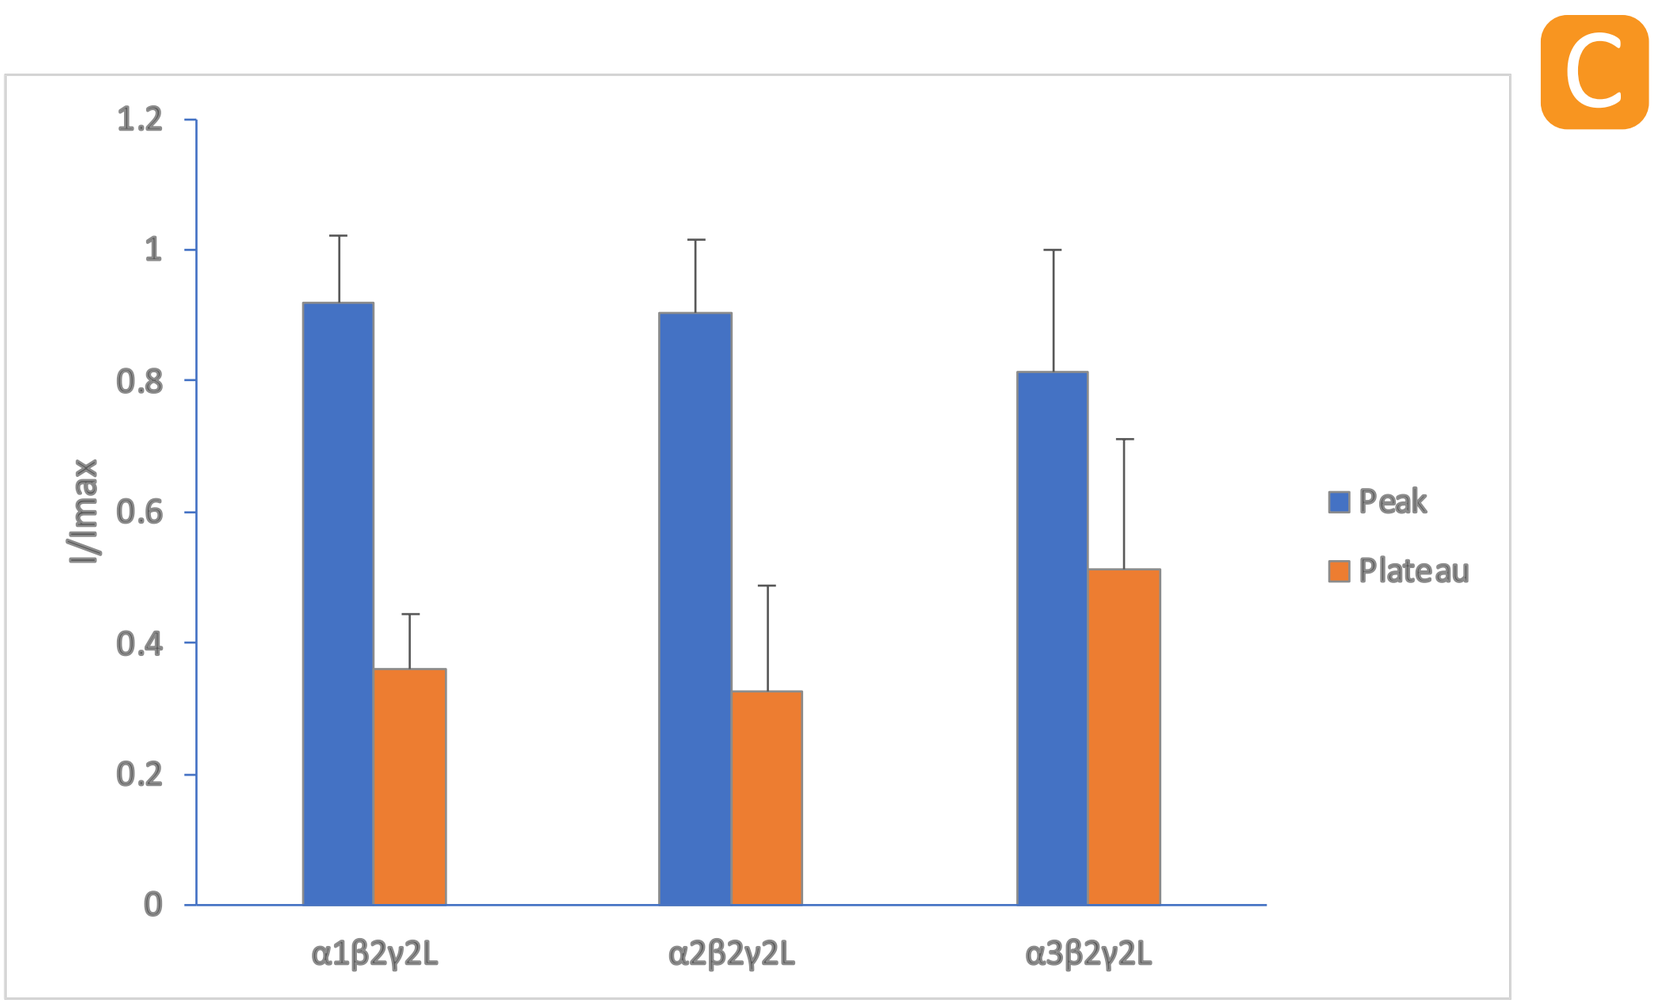

C
I/Imax
Peak
Plateau
α1β2γ2L
α2β2γ2L
α3β2γ2L

Zn^{2+} effect was measured after an incubation of 30 seconds with 30 µM $ZnCl_2$ followed by 1 mM GABA + 30 µM $ZnCl_2$. **A:** Representative superimposed serial current sweeps showing alternating effect of GABA and GABA+$ZnCl_2$. Red traces are GABA alone (Before and after wash), blue traces are GABA+ $ZnCl_2$. **B & C:** Peak and plateau maximal current under zinc baseline adjusted to current without Zn^{2+}.

Summary

With IonFlux technology, we were able to validate 6 different cell lines expressing α(1/2/3)β2γ2(L/S) GABAA receptors. We could not observe any significant differences between the L and S subunits when it came to GABA, picrotoxin and diazepam responses. Rc-Ome was shown in previous studies to have significant potentiating effect on α2β2γ2S subunits compared to α2β2. In our study we saw a potentiating effect on all subunits. Due to solubility issues we could not extrapolate the EC_{50} values for Rc-Ome. Zn^{2+}, which was also shown in other studies to have a separating effect on the subtypes, did exhibit distinctive inhibition characteristics. S subtypes showed around 50% peak and plateau inhibition with Zn^{2+} while L subtypes showed very little peak inhibition but with a significant plateau decrease.