Enhanced Control for Liquid Displacement Facilitates Assays for Positive Modulation

Ali Yehia[1], Haiyang Wei[2], Michael Donio[2]

[1]Fluxion Biosciences, South San Francisco, CA, USA, [2]Eurofins Pharma Bioanalytics Services US Inc., St Charles, MO, USA

Abstract

Ion channel modulation is an important target for drug discovery. Common compounds for ion channel activity are positive allosteric modulators (PAMs), which by binding to the protein allosteric binding site can effectively enhance channel activity while in the presence of the agonist. The activity of the modulator is considered to be in direct relationship with its concentration, provided a standardized agonist baseline effect can be had with a constant concentration of ligand. With the ability to record electrical signals across membranes of multiple cells in parallel sites, automated patch clamp systems can record the activity of a channel's agonist or antagonist in real time. However, due to the dual-channel binding and the need for repeatability of ligand response, screening for new PAMs is a delicate process, sometimes requiring quick additions of multiple compounds at the same recording site to gauge a proper comparison. Here we present data from a number of different nicotinic ion channels stably expressed in commercially-available cell lines. Using a microfluidic-based automated patch clamp system capable of fast and flexible serial displacement of agonists, modulators and antagonists, we qualify procedures that can ultimately simplify the final process of screening for PAMs as well as provide extended flexibility in ligand-gated channel studies. We present data from a variety of nicotinic receptors (α1β1δε, α3β4, α4β2, α4α6β2, and α7) showing current stability, plate to plate reproducibility, agonists, antagonists, open channel block, PAM effects, and recovery from desensitization.

Materials and Methods

Cells expressing nAChR (Eurofins Discovery Services PrecisION® cell lines – Table 1) were cultured in medium containing DMEM/F12 glutamax, Fetal Bovine Serum, Non Essential AA, selection antibiotics at 37°C and 5% CO2. For cell isolation, flasks were first washed with 10 ml of Ca- & Mg-free PBS, followed by 5 ml of Accutase® solution (Innovative Cell Technologies), and incubated at 37 degrees for 5 minutes. The cell suspension was then spun for 120 seconds (1000 rpm) and re-suspended in SFM II for at least 15 minutes. Right before experiment, the cells were re-suspended in extracellular solution. For recording nAChR currents, cells were held a holding potential of -60 mV.

Subunit	Cell Line
α1β1δε	CYL3052
α3β4	CYL3057
α4β2	CYL3106
α4α6β2	CYL3107
α7	CYL3097

Table 1

Extracellular solution (mM): 138 NaCl, 4 KCl, 1 MgCl2, 1.8 CaCl2, 10 HEPES, 5.6 glucose, pH 7.45 with NaOH.
Intracellular solution (mM): 110 TRIS-PO4, 28 TRIS-base, 0.1 CaCl2, 2 $MgCl_2$, 4 Mg-ATP, 11 EGTA, pH 7.3 with KOH.

Sources and catalog numbers of compounds used are listed in table 2. ACh was dissolved in deionized water to make a 10mM stock solutions, and diluted serially into extracellular buffer. Data analysis and graphical presentation were performed using a combination of Ionflux software and Graphpad Prism. A threshold was set at –0.2nA for responses to EC20 ACh. below which values were excluded from the analysis.

Compound	Source	Catalog
Acetylcholine	Sigma-Aldrich	A6625
Nicotine	Sigma-Aldrich	36733
Epibatidine	Sigma-Aldrich	E1145
α-Bungarotoxin	Tocris	2133
Atropine	Sigma-Aldrich	A0132
DHβE	Tocris	2349
Mecamylamine	Tocris	2843
MLA	Tocris	1029
Pancuronium	Sigma-Aldrich	P1918

System Description

The IonFlux automated patch clamp system features a unique pneumatically driven in-plate movement of solutions as well as continuous flow. IonFlux plates, based on standard configurations of 96 or 384 wells can be preloaded with cells, compounds, and reagents using conventional liquid handlers or pipetters, and loaded into the system like a simple plate reader. With its advanced software control, the system provides enhanced flexibility in fluid displacement ultimately required for proper ligand-gated assays.

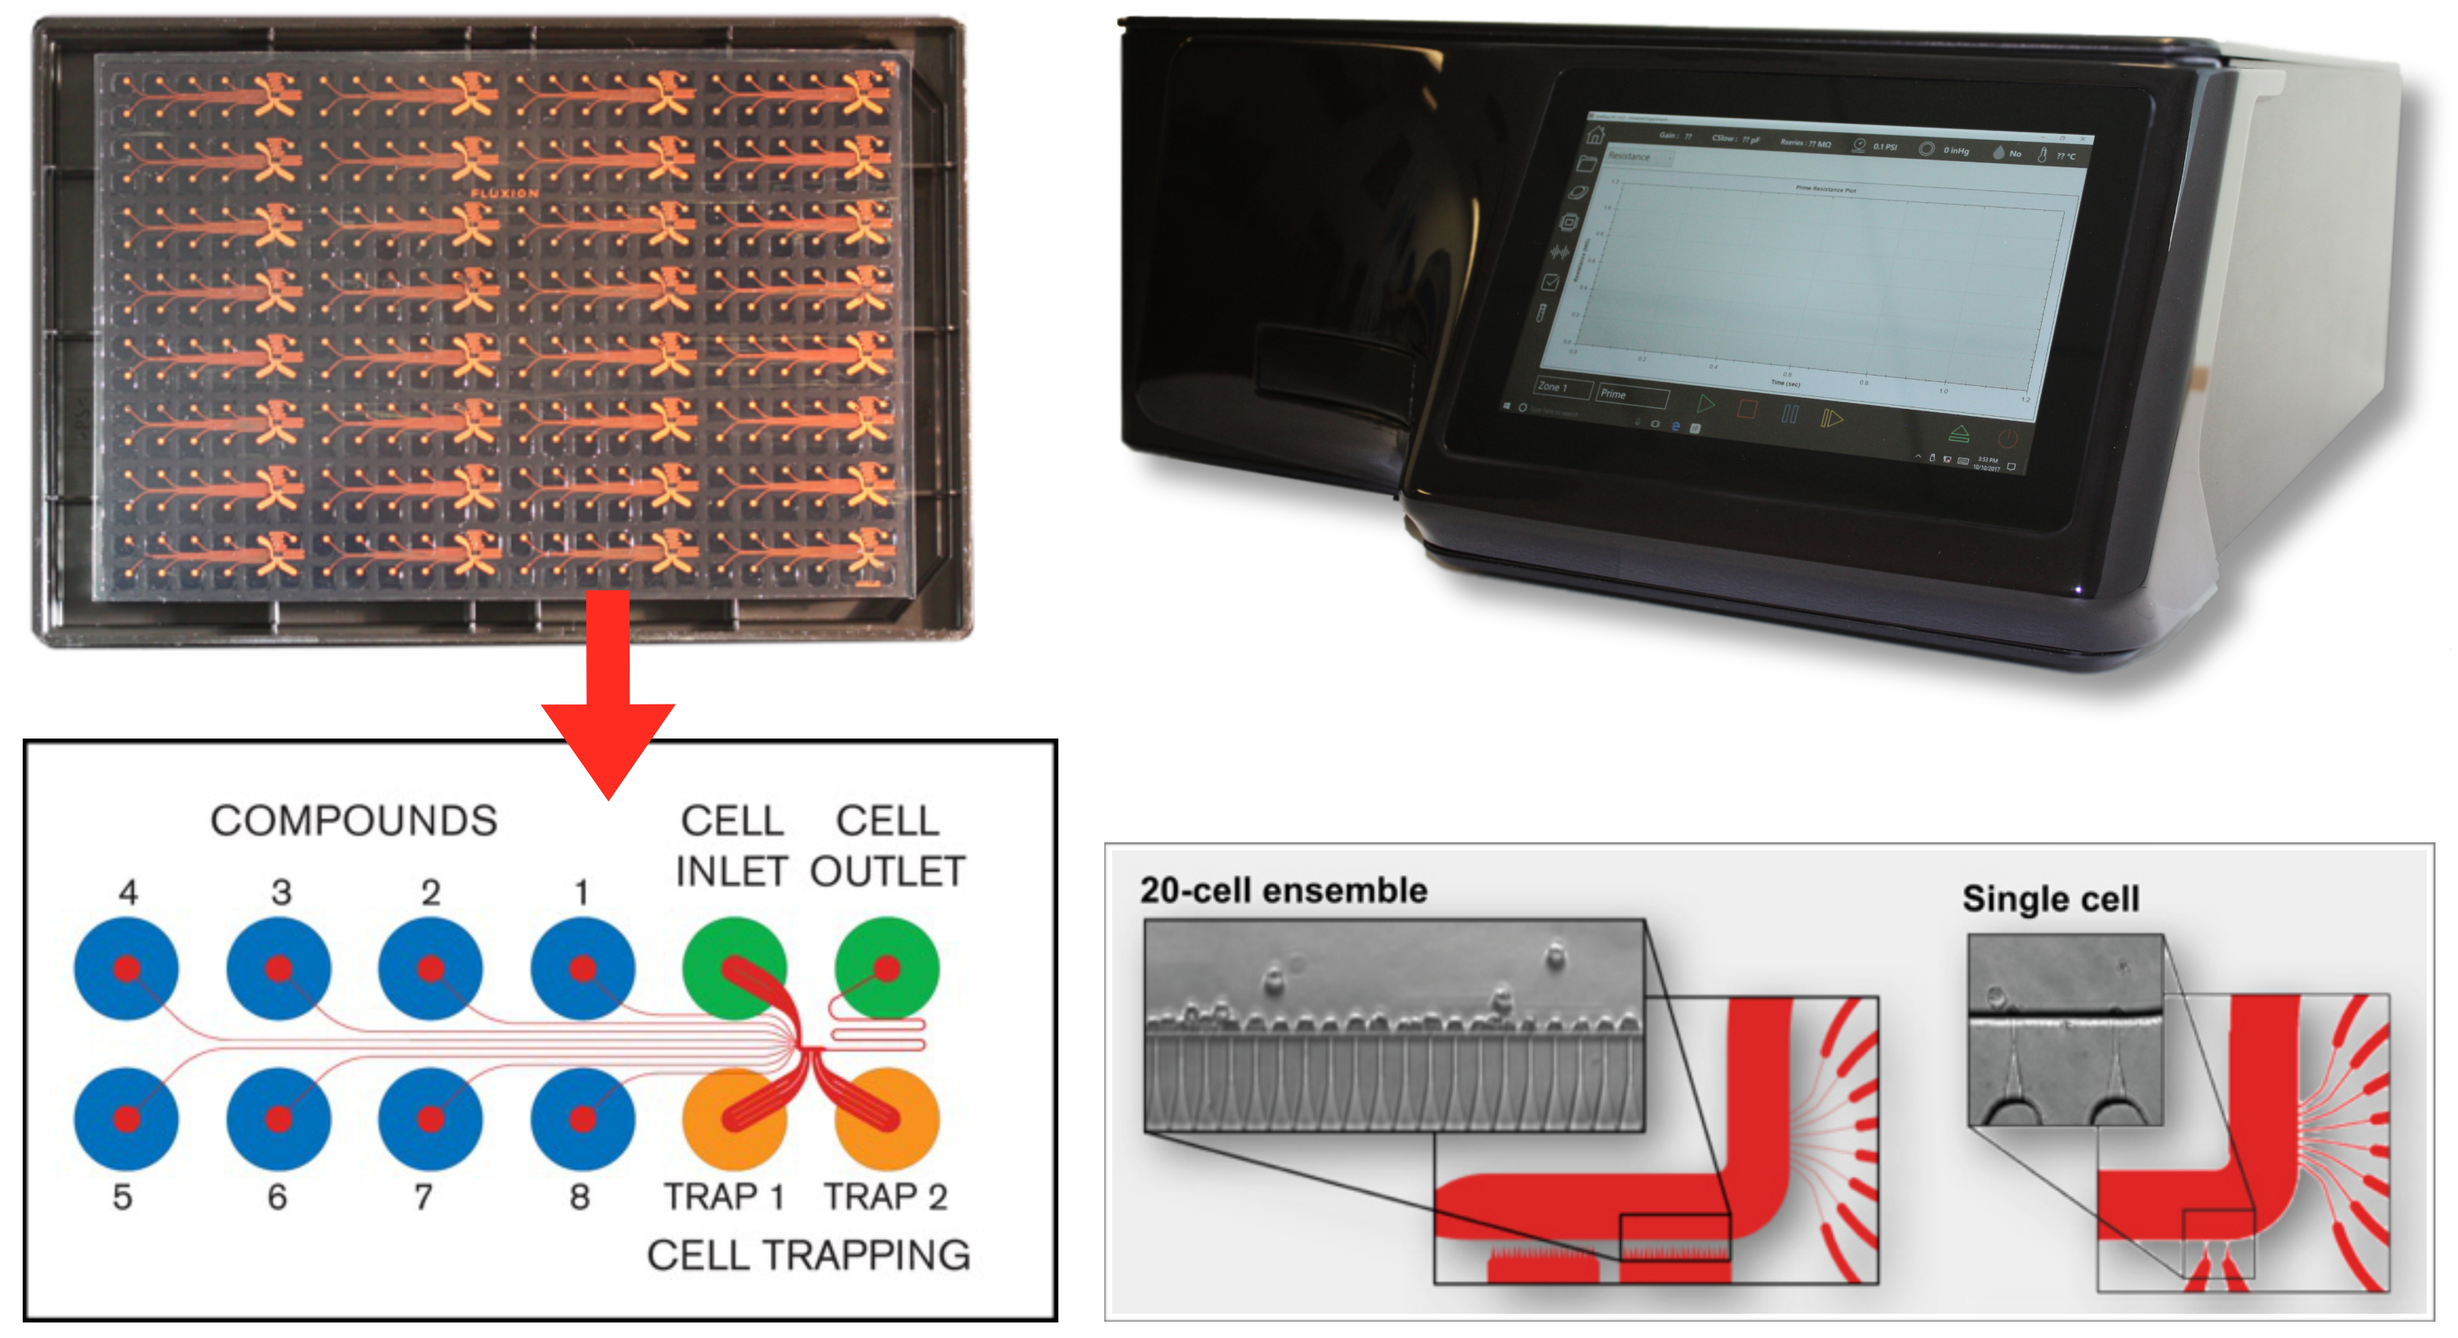
COMPOUNDS
CELL INLET
CELL OUTLET
TRAP 1
TRAP 2
CELL TRAPPING
20-cell ensemble
Single cell

Each pattern in an IonFlux plate has 8 compound wells, 2 trap recordings filled with intracellular fluid, 1 inlet for cells and extracellular solution, and 1 outlet for waste (left). Cells are captured from suspension and then patched in whole cell configuration. Plates come in two configurations: 20-cell ensemble and single cell (bottom right). Both plate formats provide high quality recordings (with giga-seals recorded in single-cell plates)

Stability

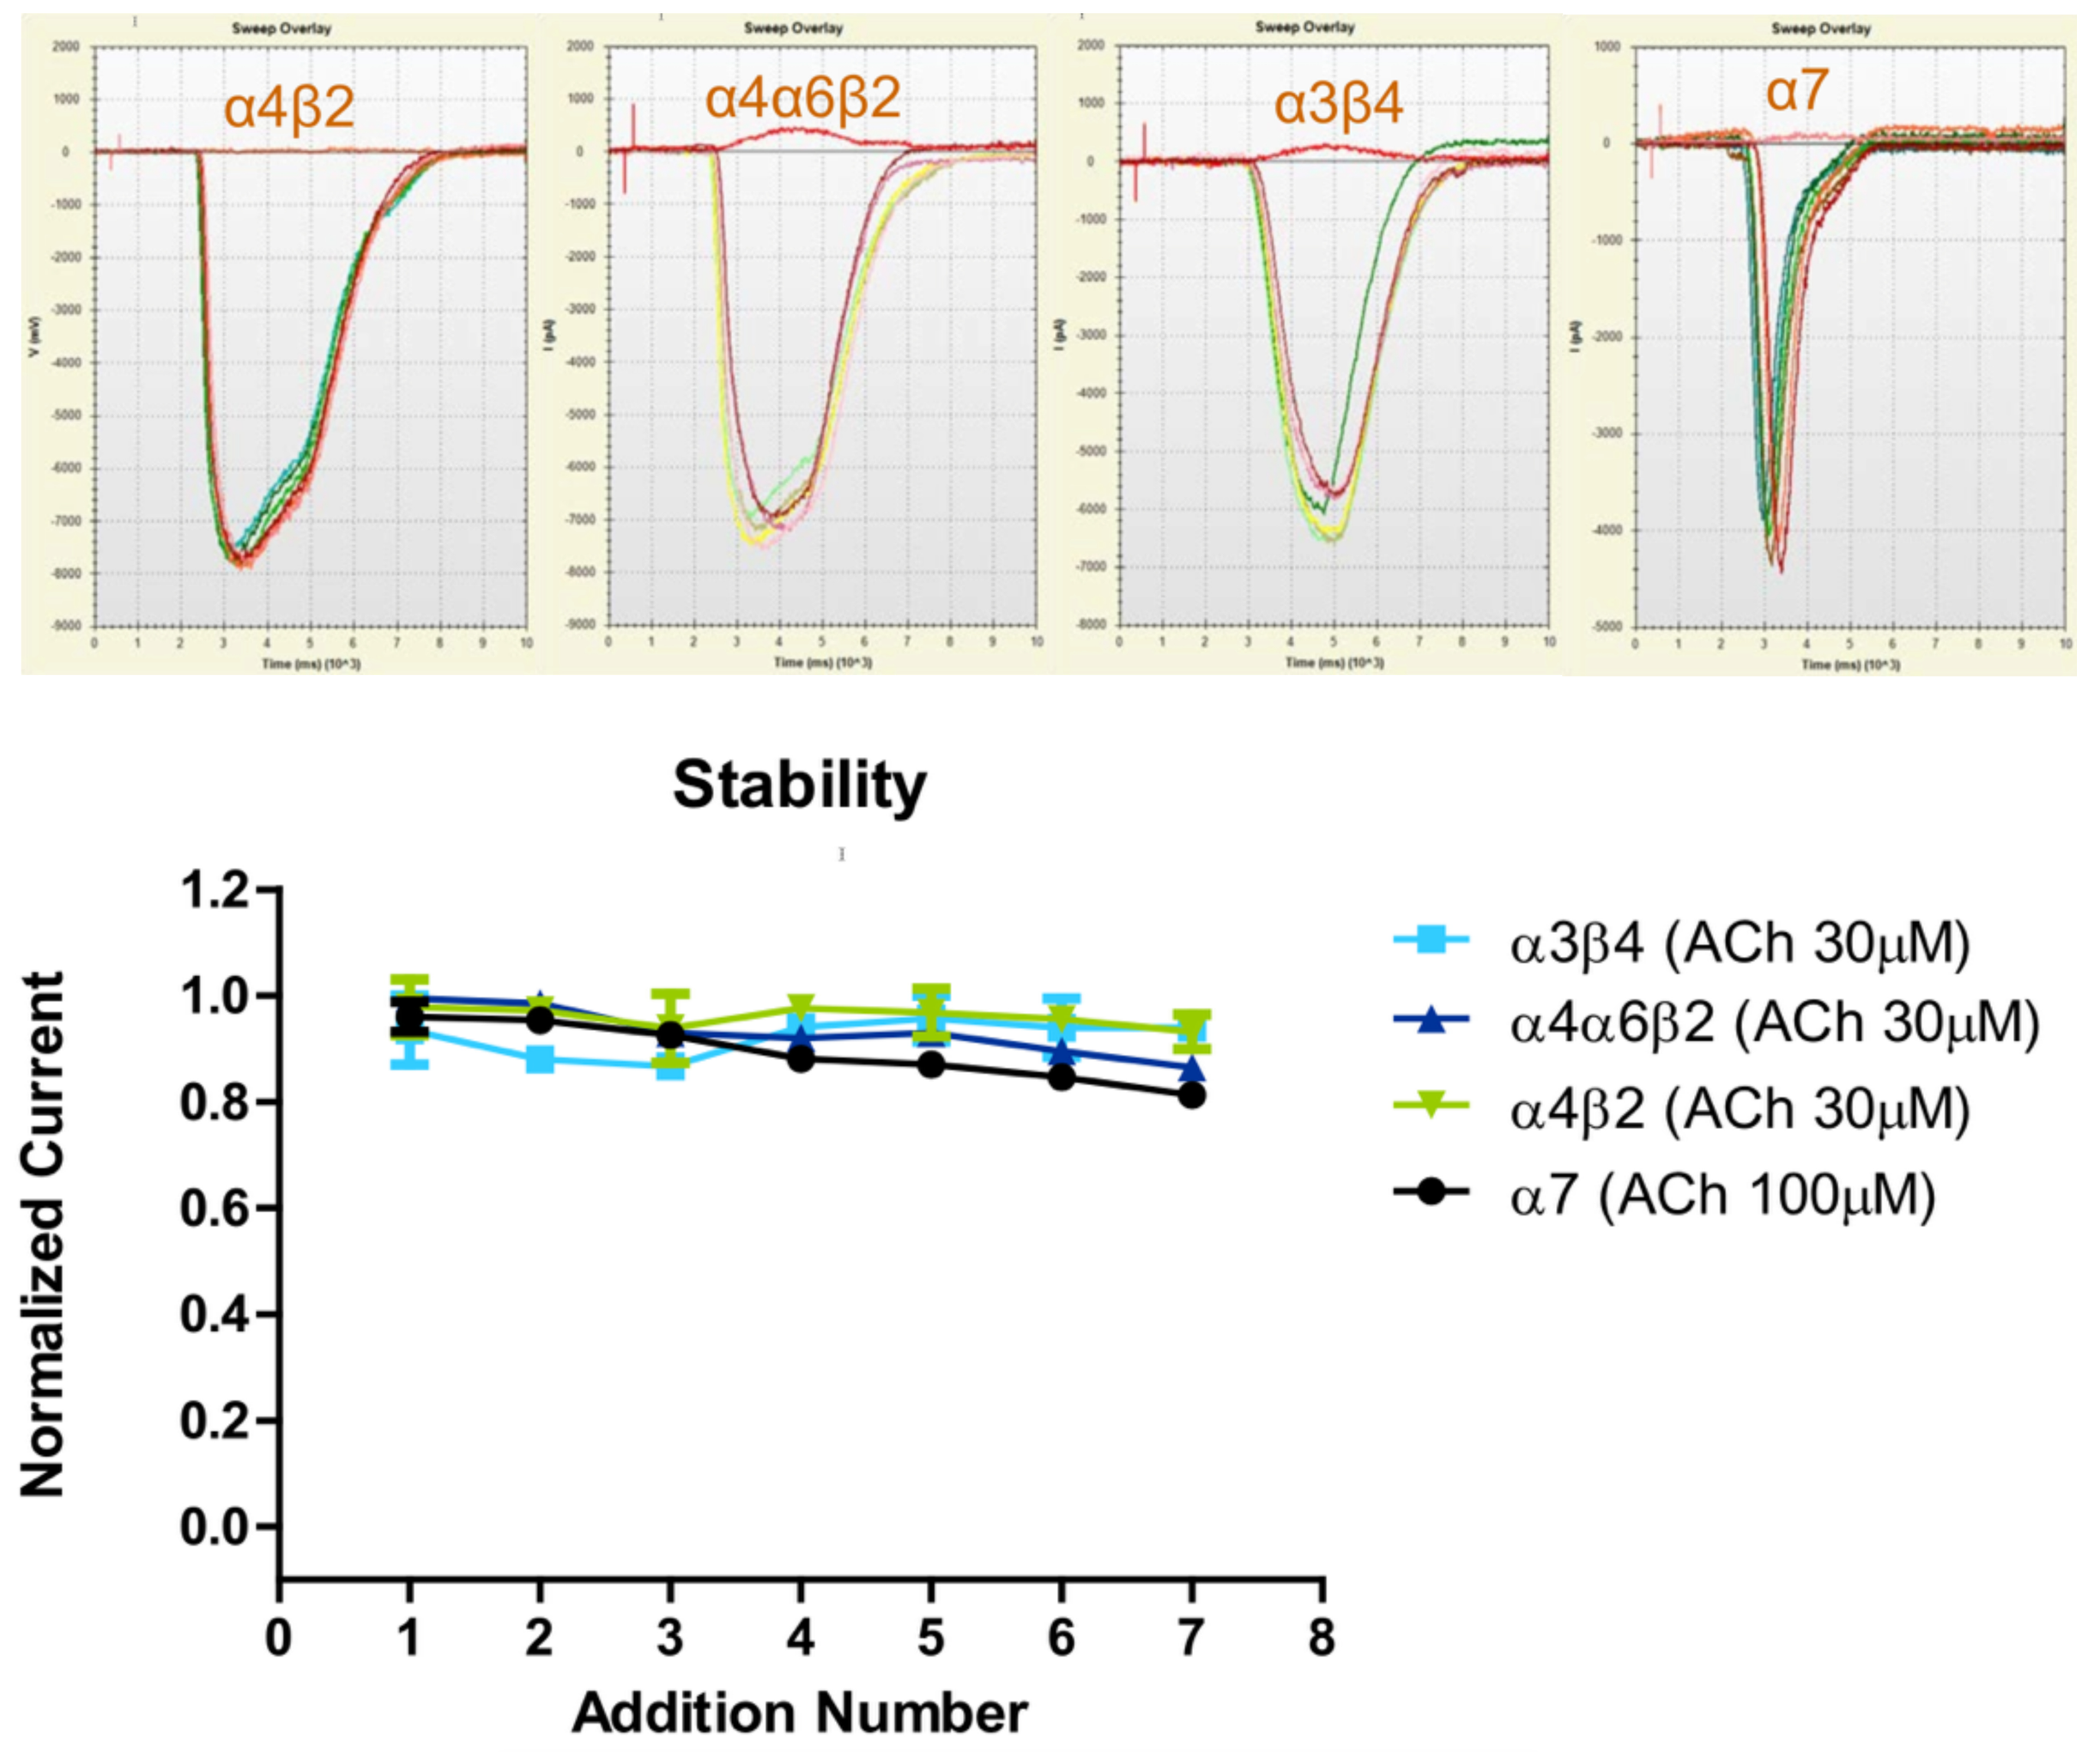
α4β2
α4α6β2
α3β4
α7
Stability
Normalized Current
Addition Number
α3β4 (ACh 30μM)
α4α6β2 (ACh 30μM)
α4β2 (ACh 30μM)
α7 (ACh 100μM)

Figure 1: Current stability was tested on repeating applications of 30 μM ACh with α4β2, α4α6β2, α3β4, and 100μM ACh with α7. Superimposed traces showing currents initiated by serial addition of ACh (top). Plotting normalized values of maximal current from the different cell lines shows minimal variability and run-down in current magnitude (bottom).

Reproducibility

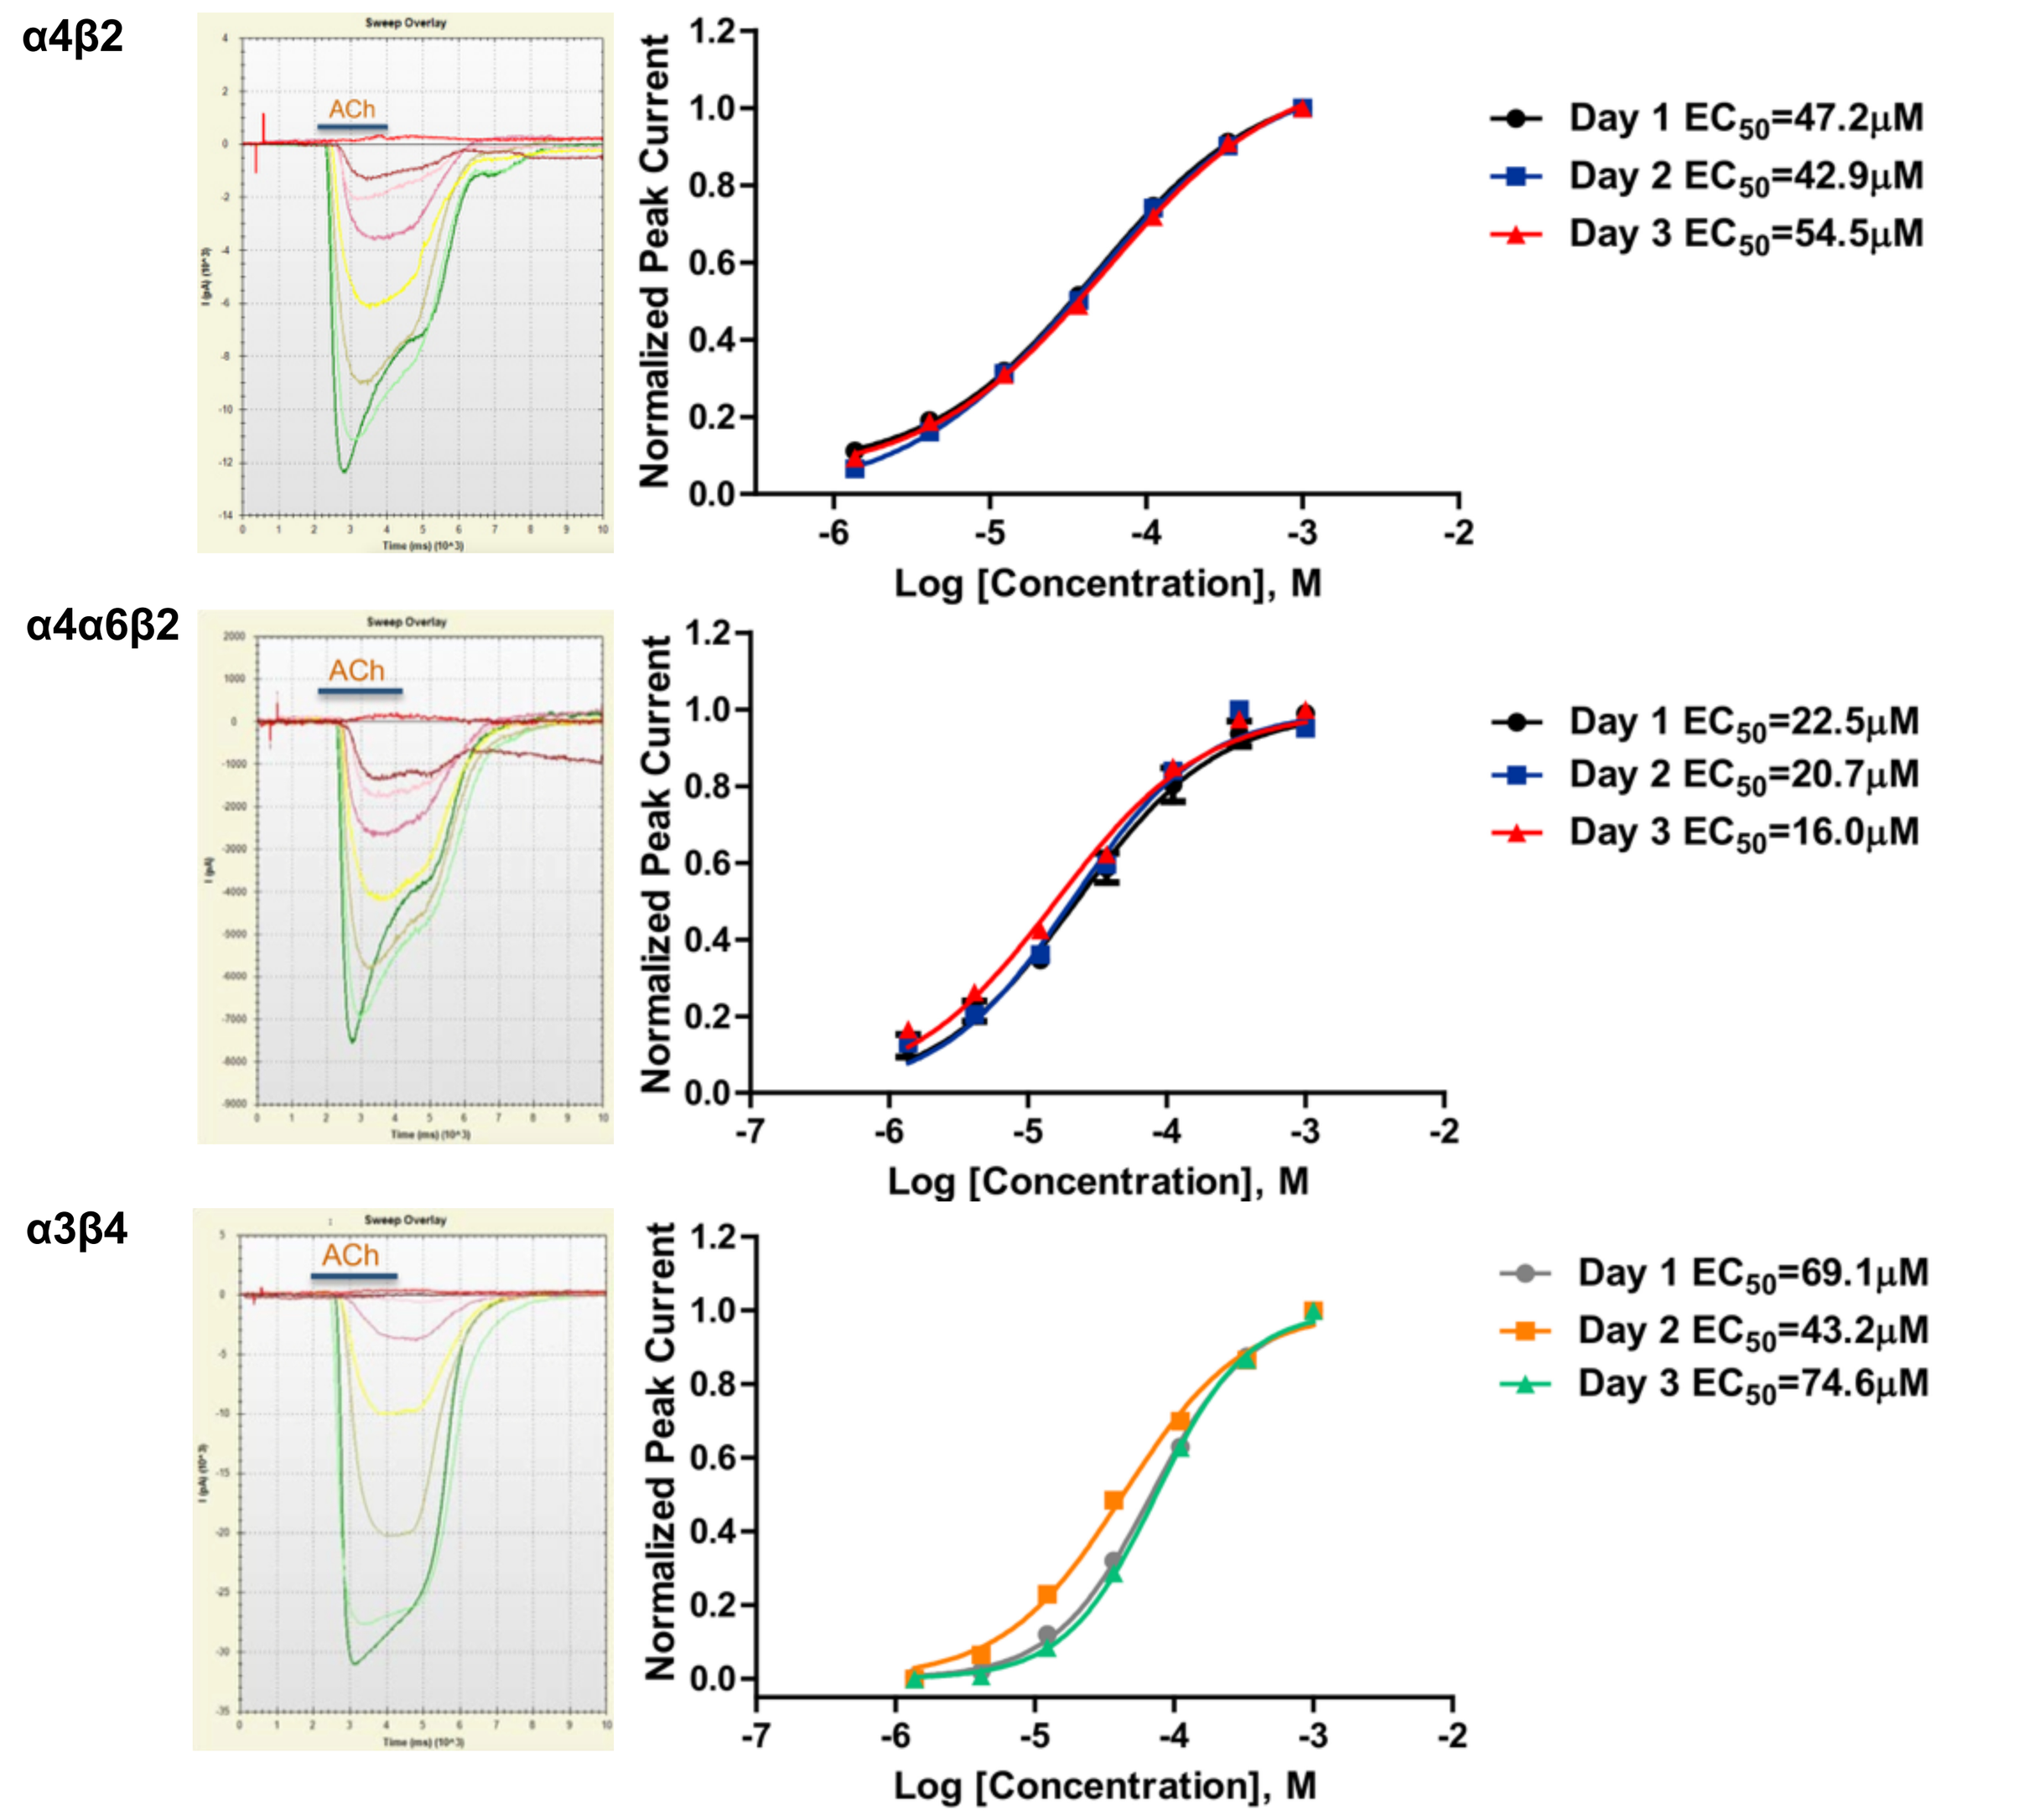
α4β2
ACh
Normalized Peak Current
Log [Concentration], M
Day 1 EC50=47.2μM
Day 2 EC50=42.9μM
Day 3 EC50=54.5μM
α4α6β2
Day 1 EC50=22.5μM
Day 2 EC50=20.7μM
Day 3 EC50=16.0μM
α3β4
Day 1 EC50=69.1μM
Day 2 EC50=43.2μM
Day 3 EC50=74.6μM

Figure 2: Normalized dose response curves for α4β2, α4α6β2, and α3β4 over three days of experiments. The day-to-day as well as plate-to-plate variability was minimal showing high reproducibility of results.

Agonists & Antagonist

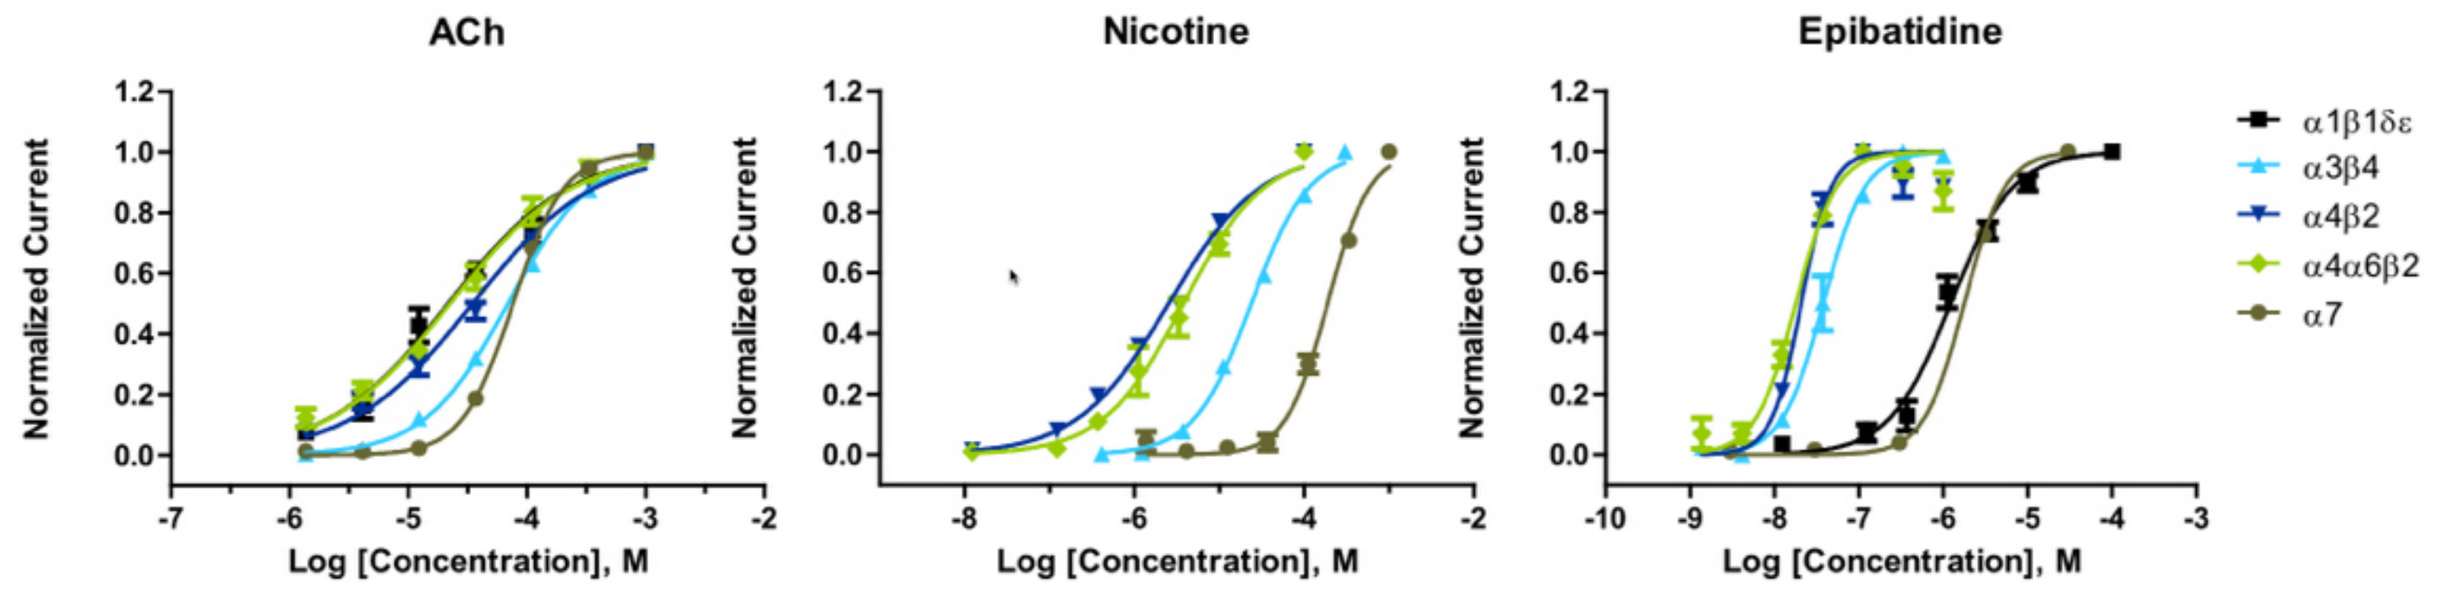
ACh
Nicotine
Epibatidine
Normalized Current
Log [Concentration], M
α1β1δε
α3β4
α4β2
α4α6β2
α7

Figure 3: Agonists> ACh, Nicotine and Epibatidine dose response curves with α1β1δε

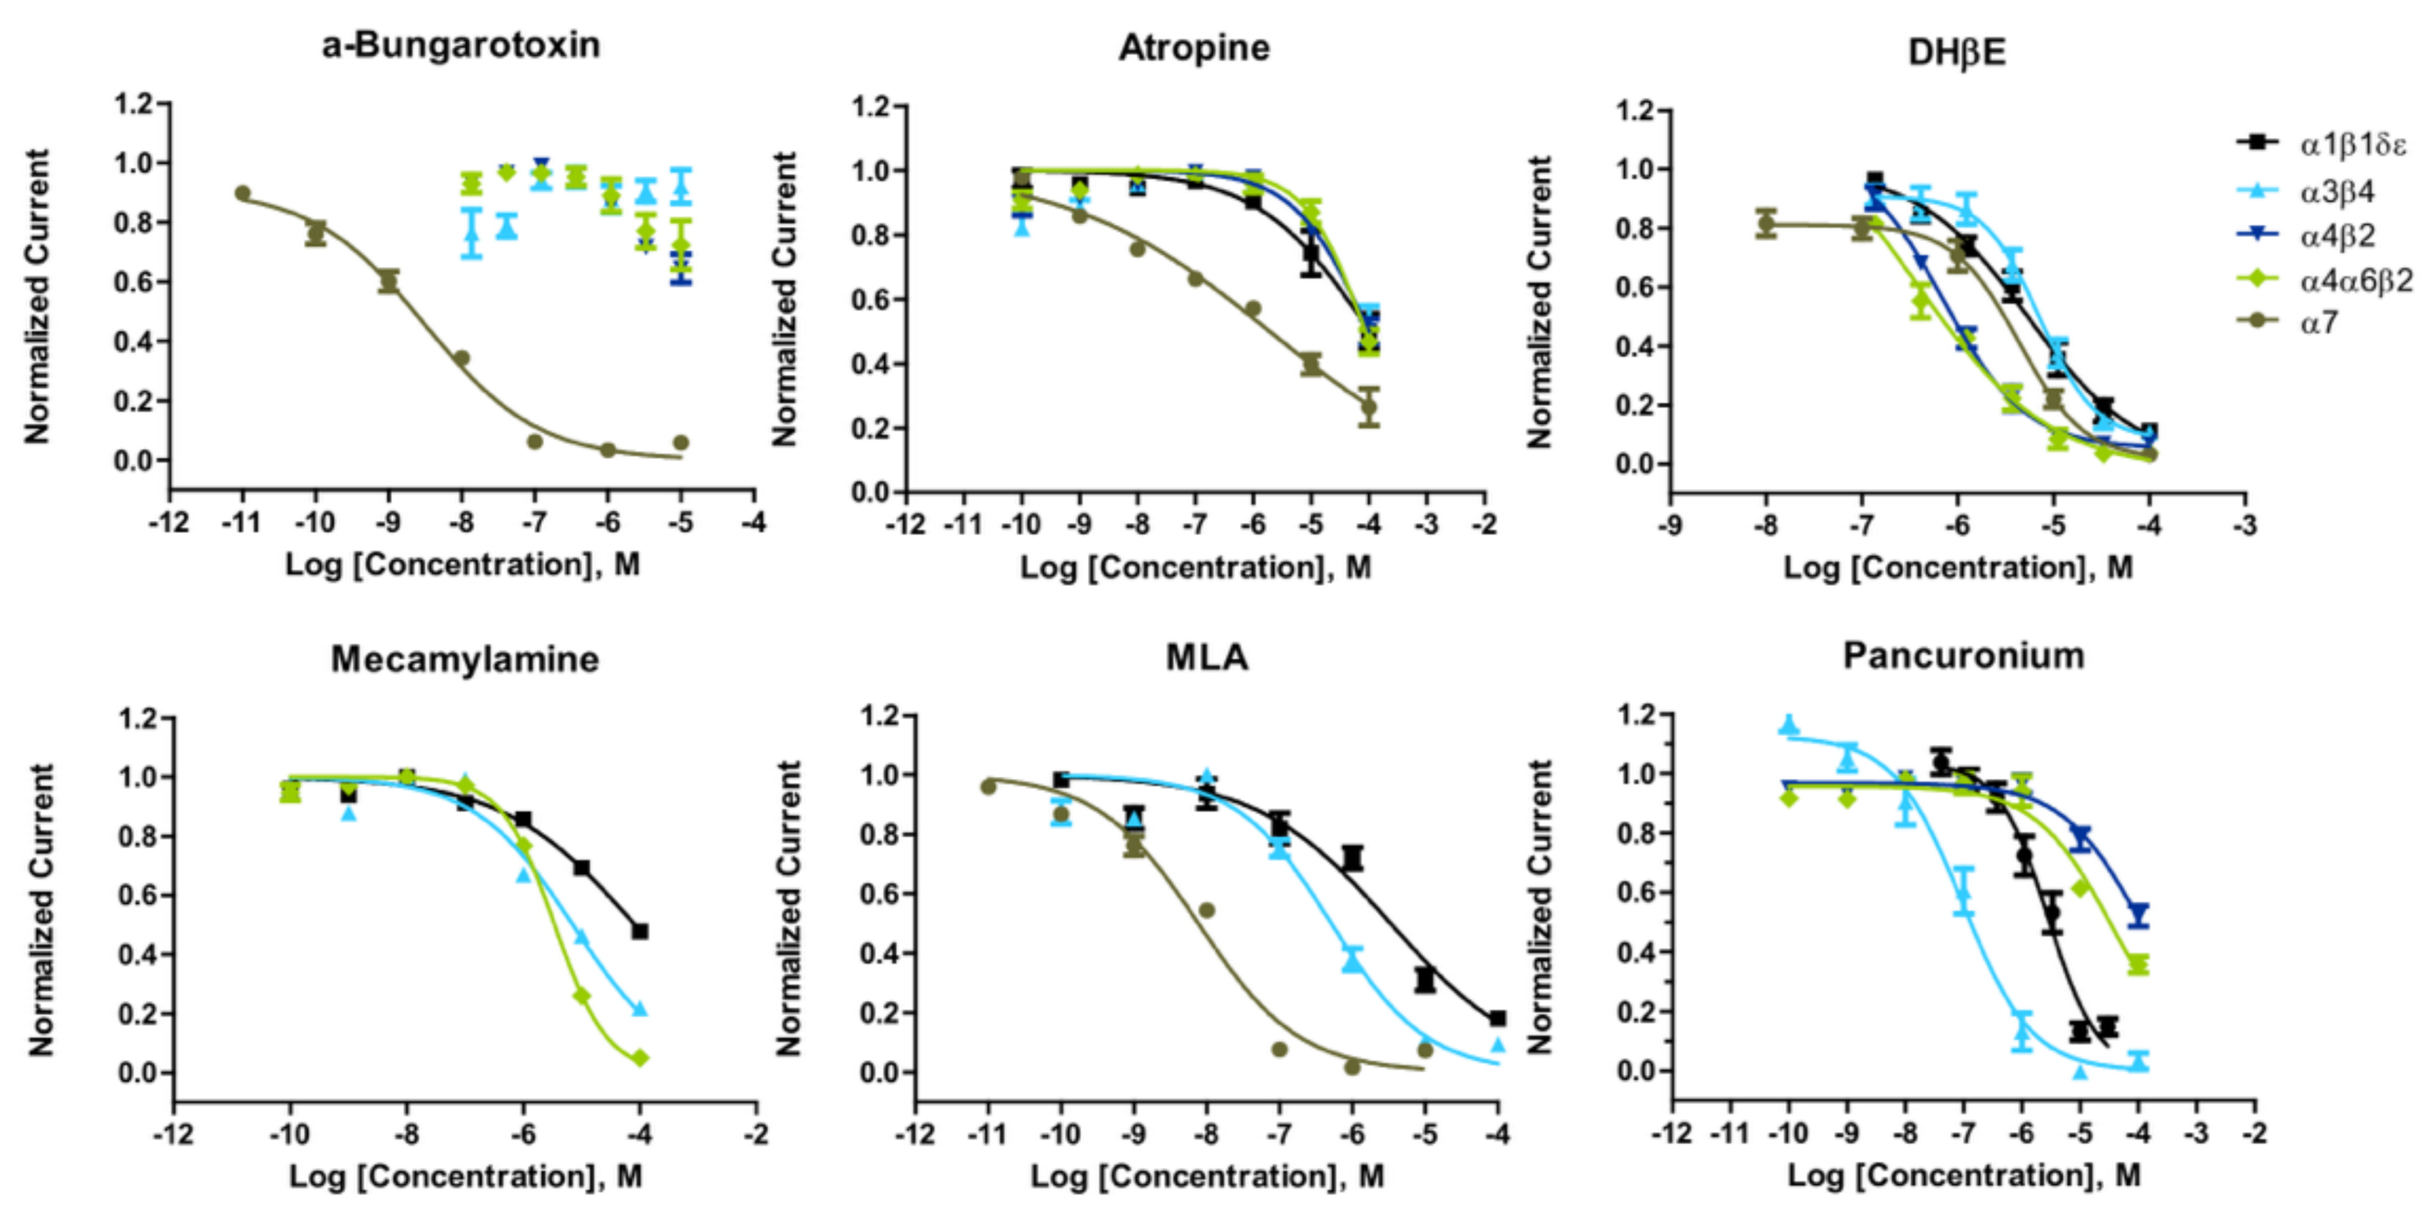
a-Bungarotoxin
Atropine
DHβE
Mecamylamine
MLA
Pancuronium
Normalized Current
Log [Concentration], M
α1β1δε
α3β4
α4β2
α4α6β2
α7

Figure 4: Antagonists> α-Bungarotoxin, Atropine, DHβE, Mecamylamine, MLA, Pancuronium dose response curves with α1β1δε, α3β4, α4β2, α4α6β2 and α7.

	ACh	Nicotine	Epibatidine	α-BTX	Atropine	DHβE	Mecamylamine	MLA	Pancuronium
α1β1δε	21.8	-	1.23	-	103.6	5.4	82.4	3.3	2.8
α3β4	69.1	23.9	0.0378	-	-	7.2	7.0	0.5	0.096
α4β2	34.9	2.54	0.0201	-	99.0	0.7	-	-	119.7
α4α6β2	22.5	3.79	0.0179	-	86.7	0.4	3.4	-	36.9
α7	76.4	188.4	1.77	0.0032	1.9	4.3	-	0.0077	-

Table 3: EC_{50} and IC_{50} in μM

Pharmacology of PAM

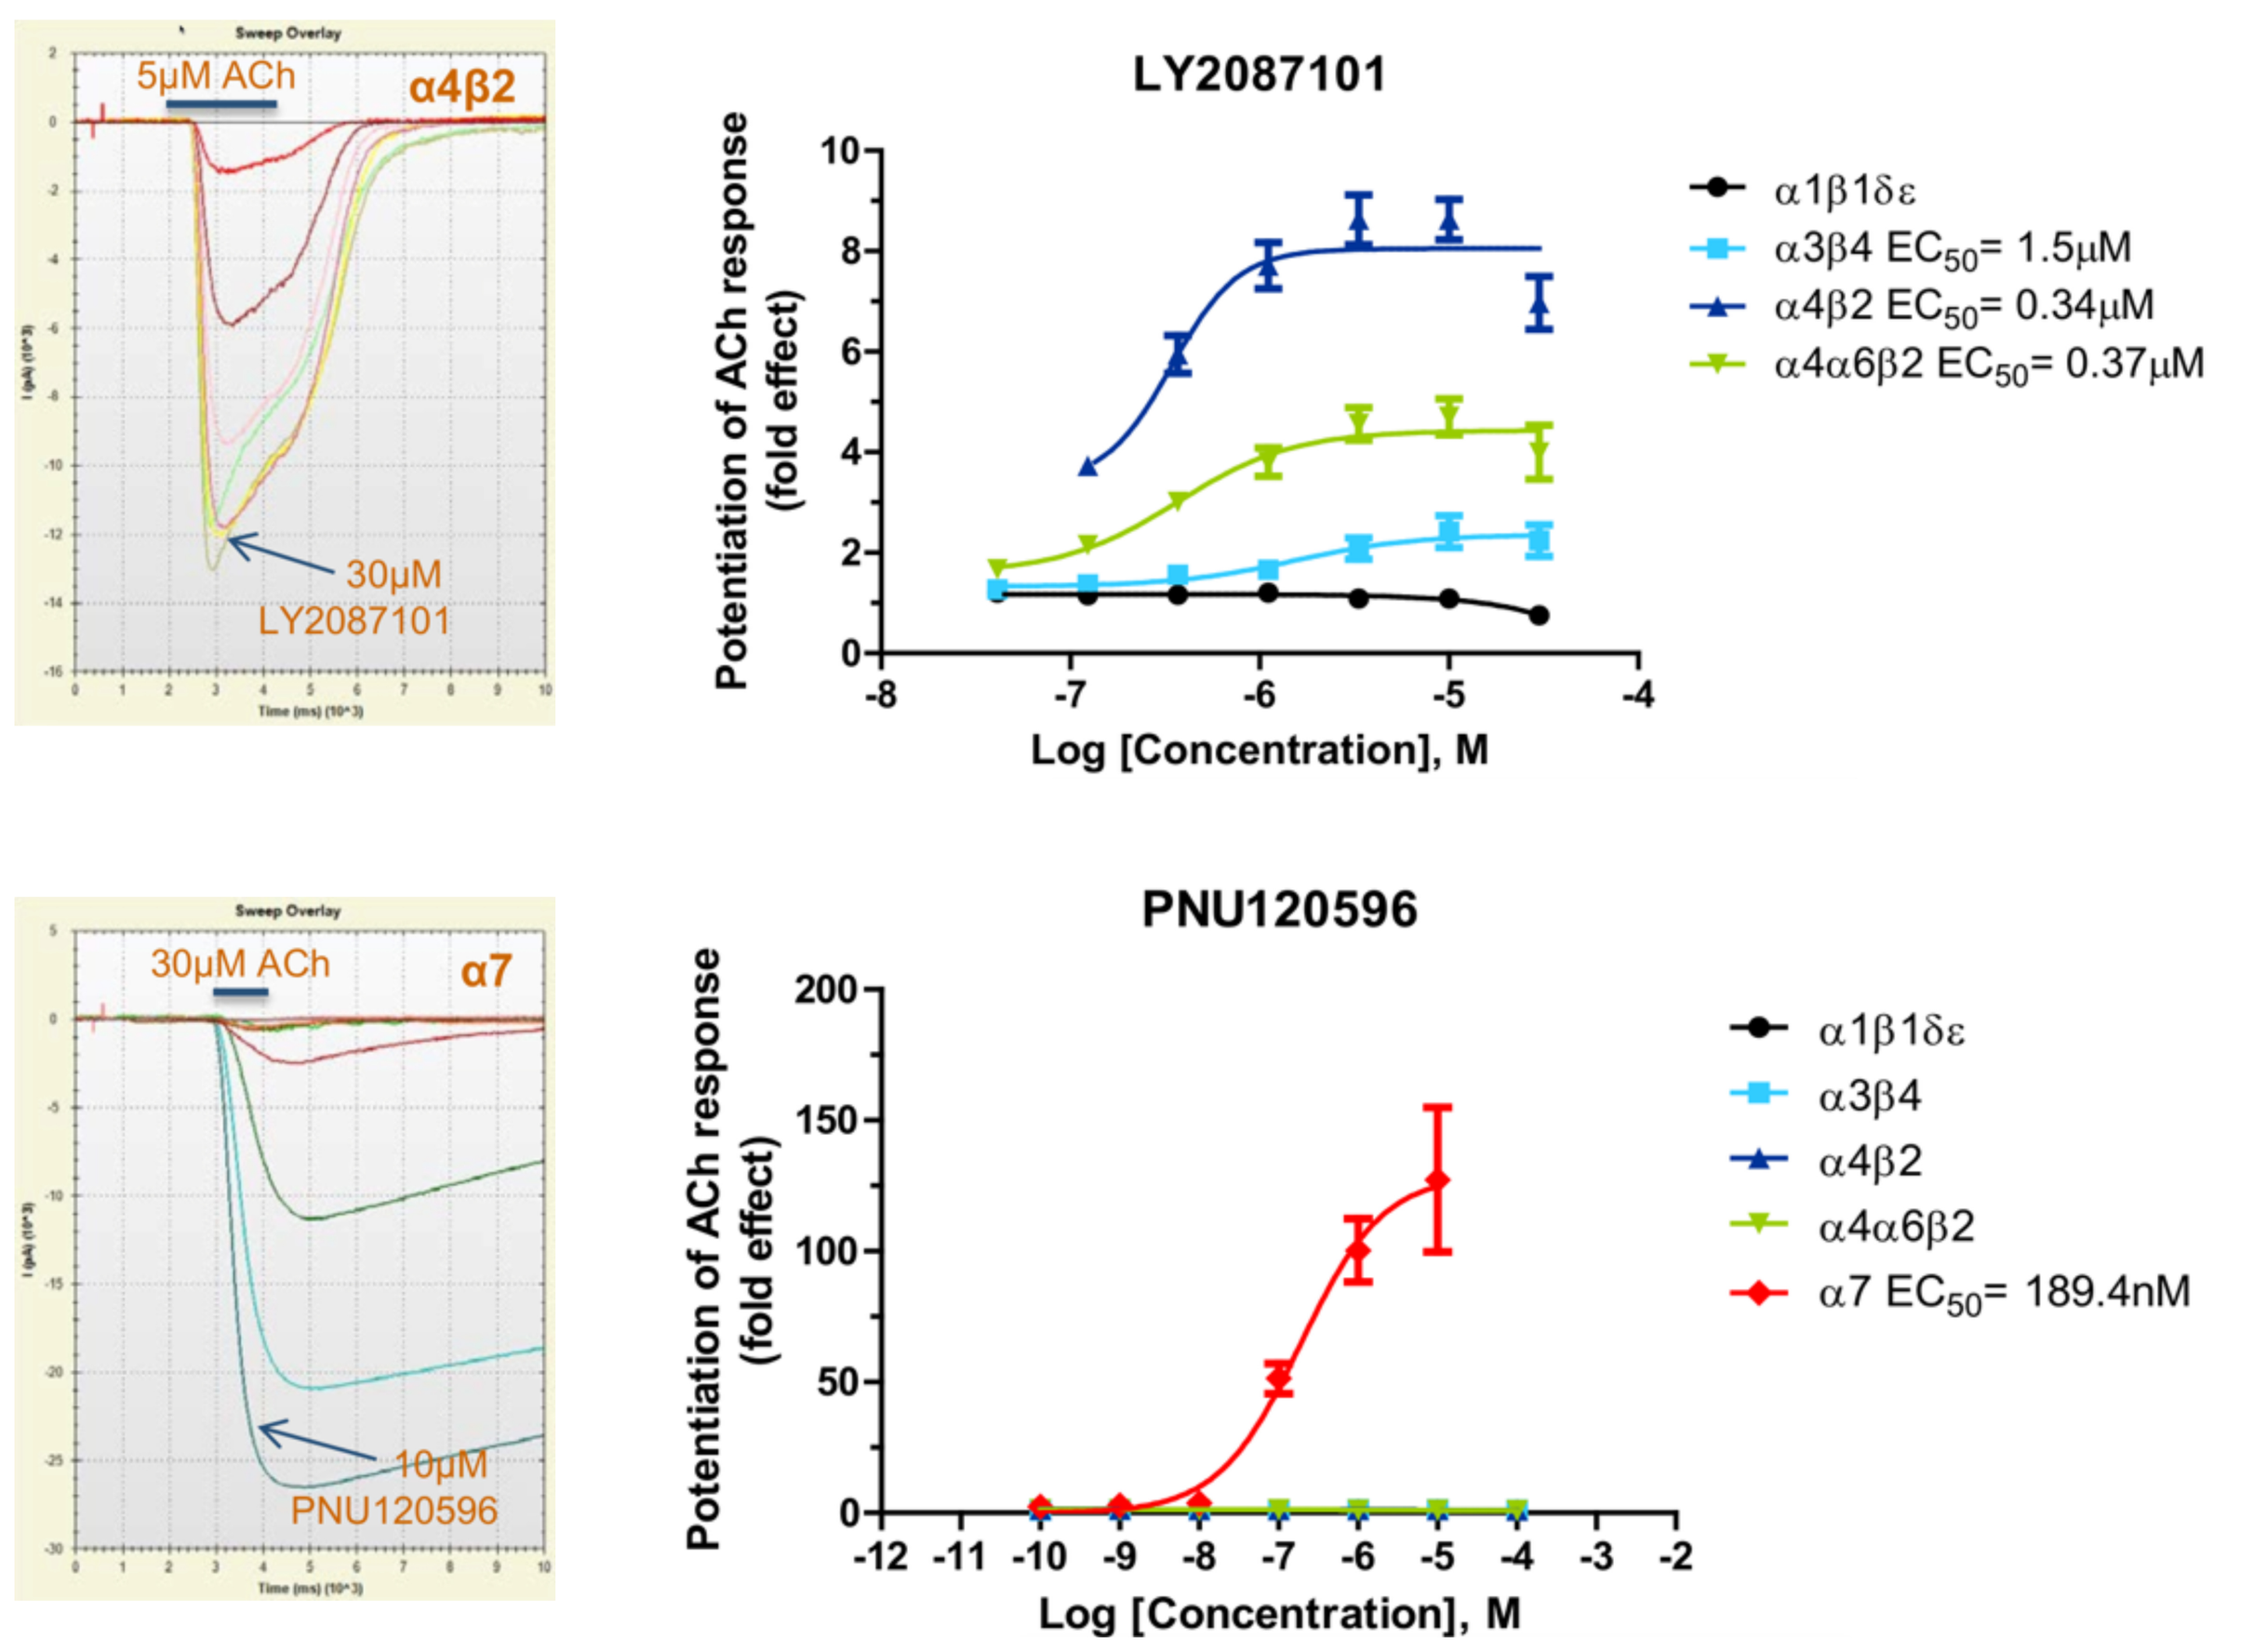
5μM ACh
α4β2
30μM
LY2087101
LY2087101
Potentiation of ACh response (fold effect)
Log [Concentration], M
α1β1δε
α3β4 EC50= 1.5μM
α4β2 EC50= 0.34μM
α4α6β2 EC50= 0.37μM
30μM ACh
α7
10μM
PNU120596
PNU120596
α1β1δε
α3β4
α4β2
α4α6β2
α7 EC50= 189.4nM

Figure 5: Normalized dose response curves for α1β1δε, α3β4, α4β2, α4α6β2 and α7 with LY2087101 (top) and PNU120596 (bottom). Agonist is ACh 5μM (top) and 30μM (bottom). LY2087101 showed no effect on α1β1δε and was not tried on α7. PNU120596 showed effect only on α7.

Recovery from Desensitization

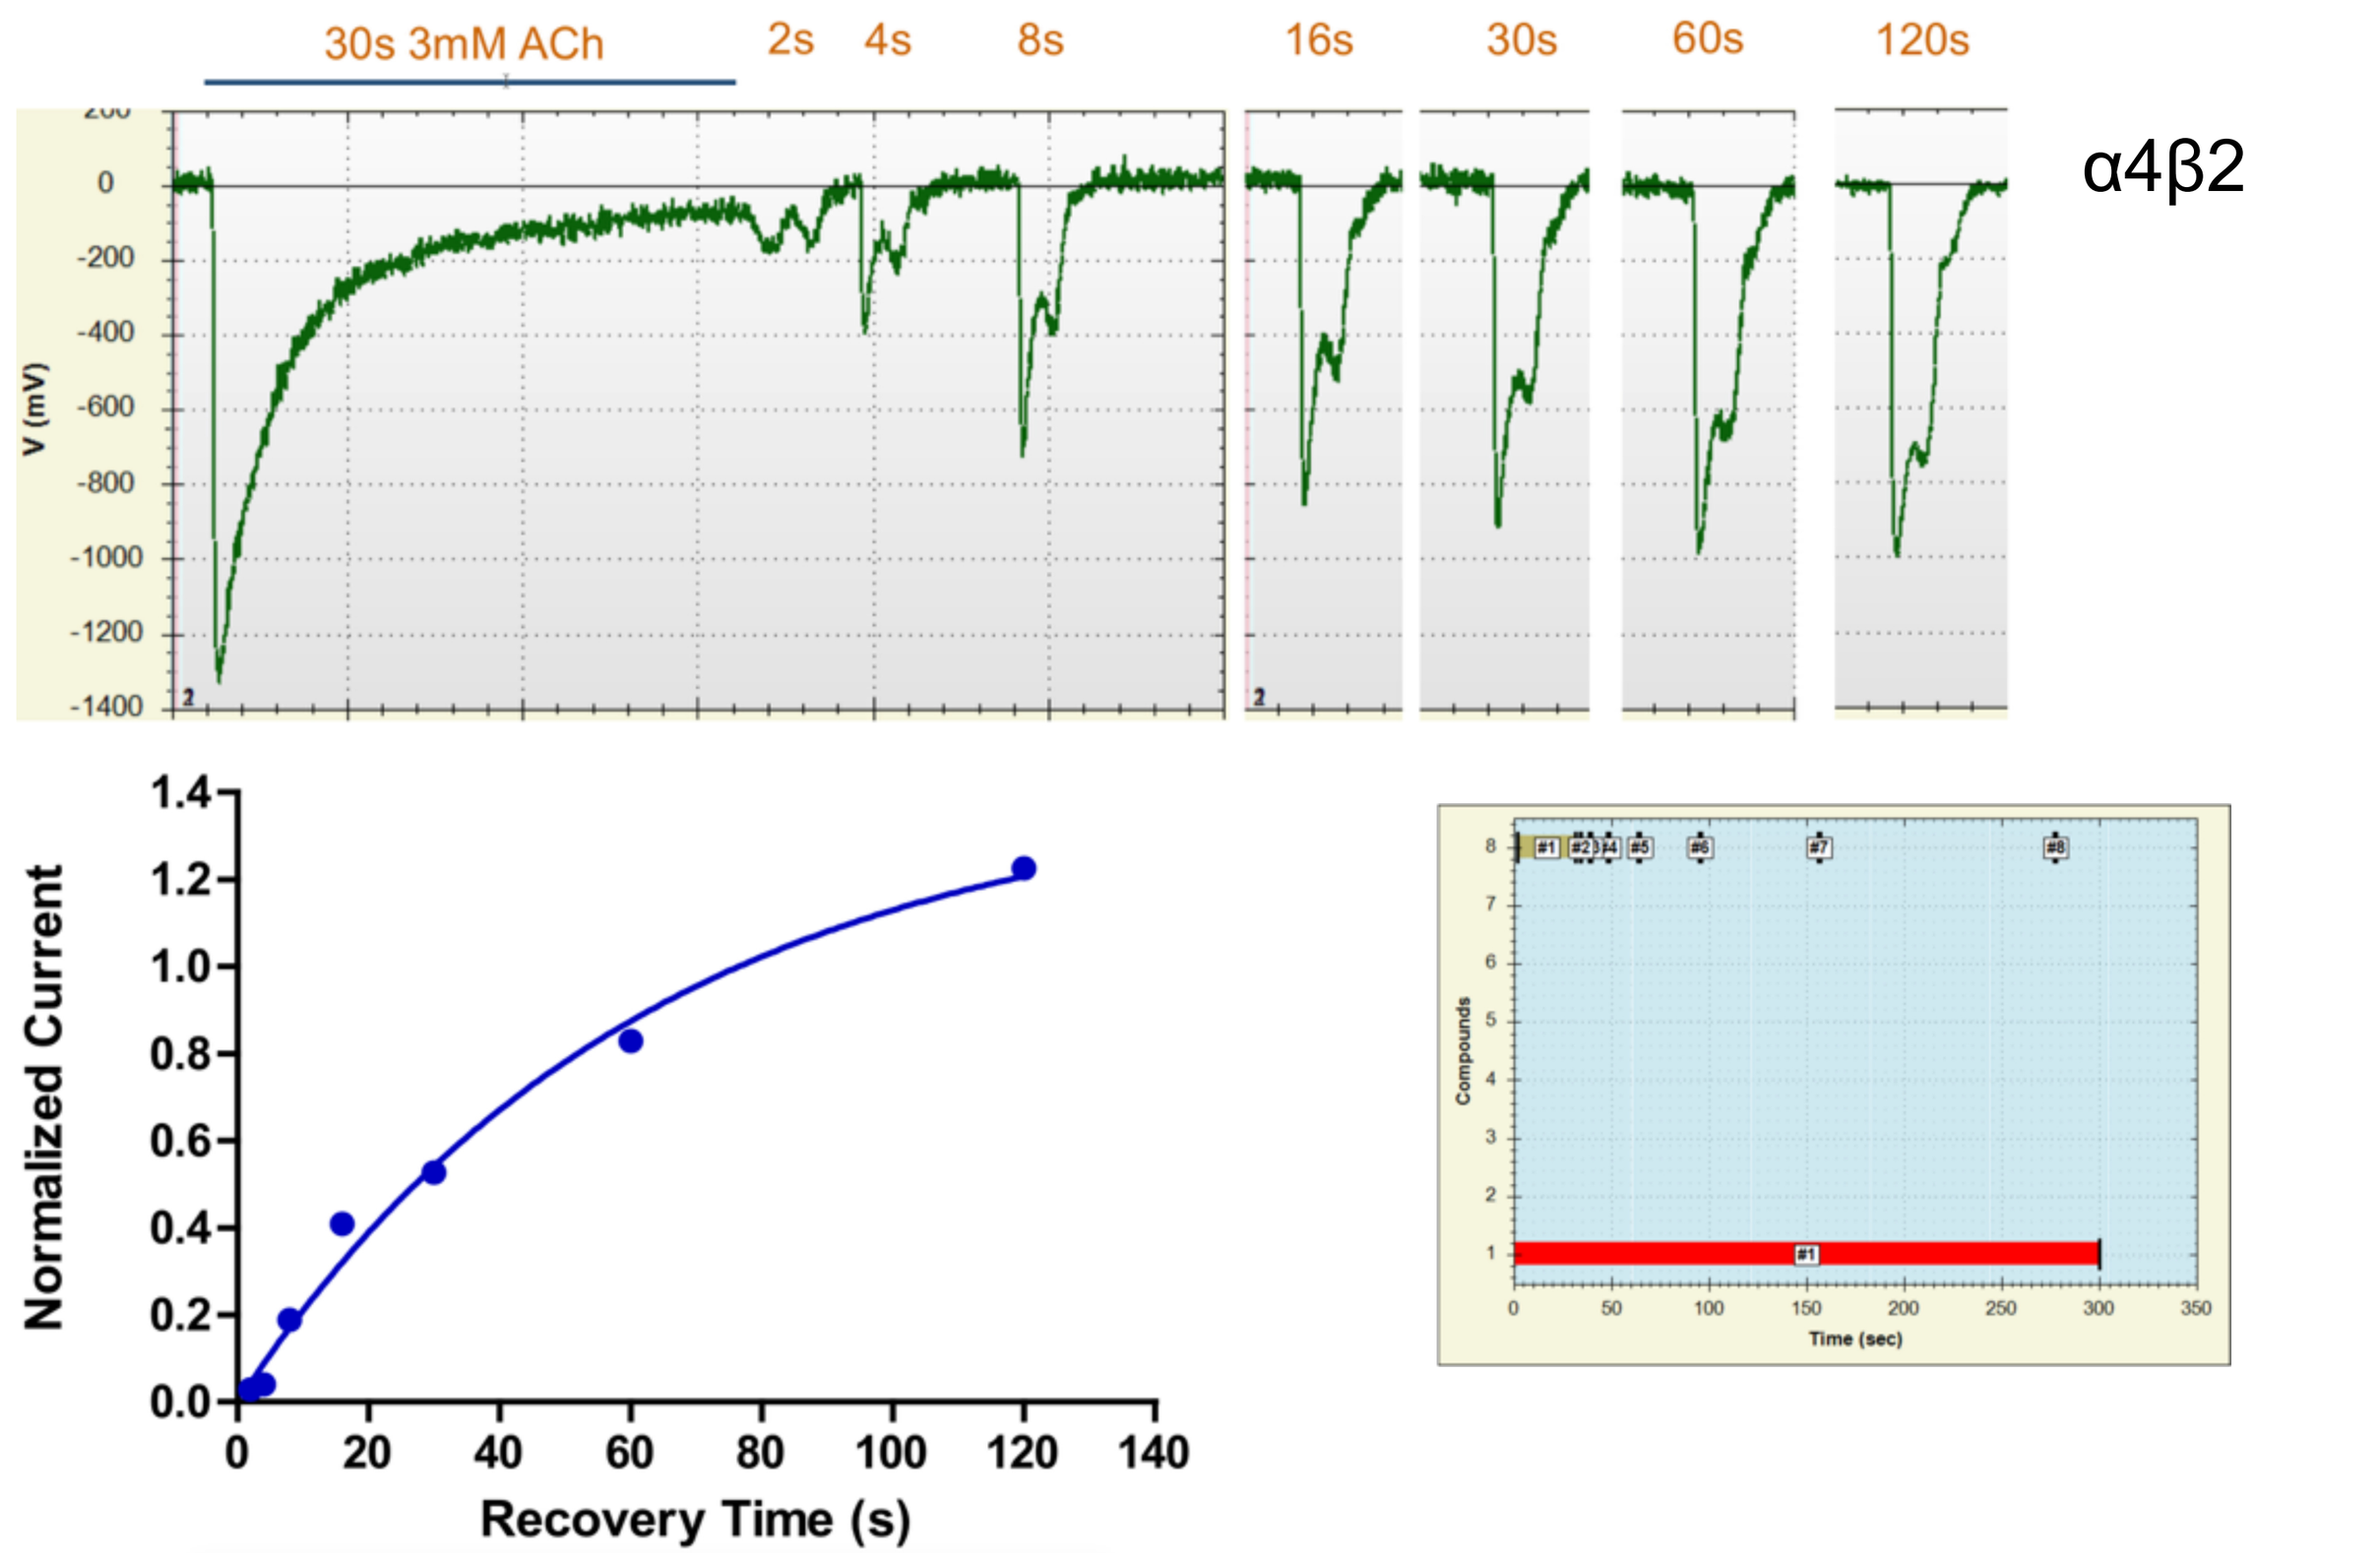
30s 3mM ACh
2s
4s
8s
16s
30s
60s
120s
α4β2
V (mV)
Normalized Current
Recovery Time (s)
Compounds
Time (sec)

Figure 6: Recovery from desensitization of the α4β2 receptor in response to 3mM ACh after increasing wash periods. Full desensitization was done with 30 s application of Ach. Protocol design displayed bottom right. Peak normalized current plotted against recovery (wash) time showing exponential recover (bottom left)

Conclusion

Discovery and research of new effective PAMs can be a challenge for most high throughput automated patch clamp system due to intermittent buffer exchange, incomplete washout, inconsistent serial receptor response, or non-parallel execution of assays. The unique liquid exchange and continuous flow system present on the IonFlux system allows consistent peak current responses in the presence of constant serial agonist additions. This provides for easy detection and measurement of variability in receptor response when additional factors are present such as PAMs. The precision in fluid control sets the IonFlux apart providing the foundation for easy screening of complex assays.